41P

ARX517, a Next-Generation Anti-PSMA Antibody Drug Conjugate for the Treatment of Metastatic Castration-Resistant Prostate Cancer, Demonstrates Anti-tumor Activity in Enzalutamide-Resistant and Enzalutamide-Sensitive Models and a Clear Therapeutic Index in a Non-human Primate Model

L. Skidmore, D. Mills, J-Y. Kim, P. Shastri, N. A. Knudsen, J. Steen, J. Nelson, Y. Buechler, F. Tian, S. Aung, S. Zhang

Ambrx, Inc.

INTRODUCTION

Enzalutamide has been approved for the treatment of metastatic castration-resistant prostate cancer (mCRPC), however patients eventually develop resistance and progress.[1] Multiple groups have also reported enzalutamide treatment can upregulate prostate-specific membrane antigen (PSMA) expression in mCRPC patients[2-4], providing strong rationale for treating enzalutamide-resistant mCRPC patients with a PSMA-targeted antibody drug conjugate (ADC).

- Previous ADCs targeting PSMA are no longer under active clinical development, likely due to ADC instability leading to unacceptable toxicity or a narrow therapeutic index in patients.
- ARX517 is a next generation anti-PSMA ADC that employs stable oxime conjugation chemistry and a non-cleavable PEG linker to ensure ADC stability and overcome the issues encountered by earlier ADCs.
- ARX517 is comprised of a humanized anti-PSMA antibody site-specifically conjugated to amberstatin 269 (AS269) drug-linker to generate an ADC with a controlled drug-to-antibody ratio (DAR) of 2.

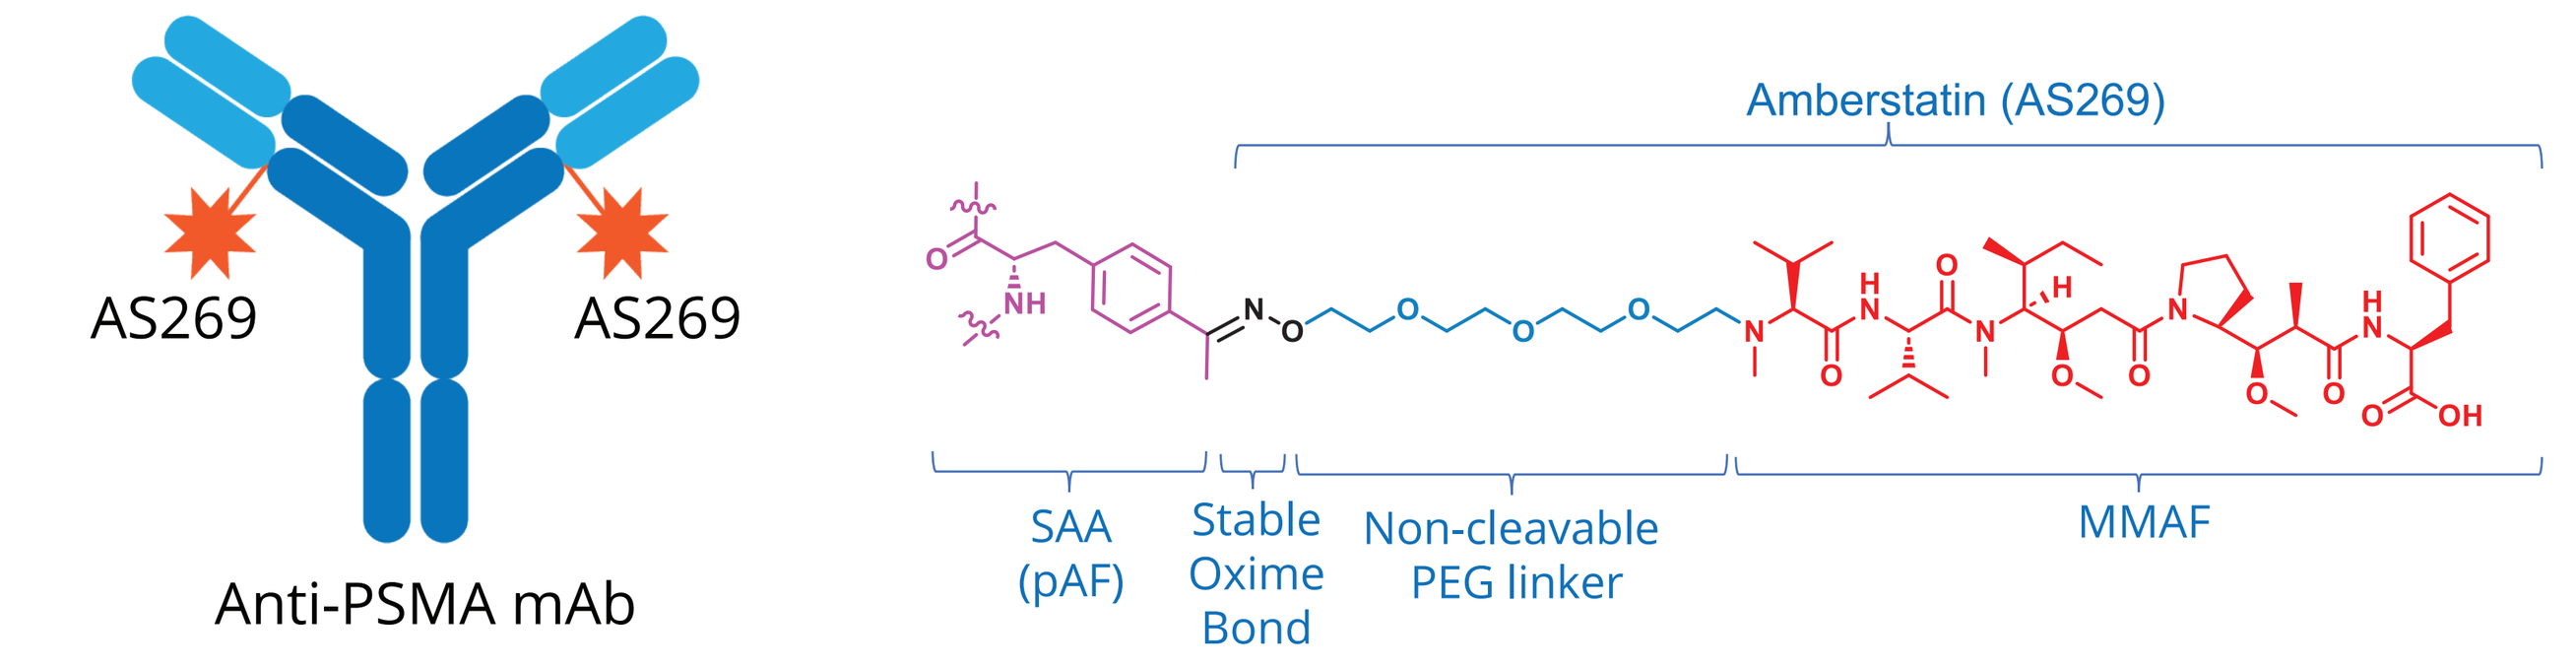
AS269
AS269
Anti-PSMA mAb
Amberstatin (AS269)
SAA (pAF)
Stable Oxime Bond
Non-cleavable PEG linker
MMAF

- Here we describe the preclinical characterization of ARX517 with in vitro and vivo efficacy, PK and stability, and therapeutic index studies.
- Preliminary FIH safety data, from an on-going Phase 1 dose escalation trial (APEX-01), show a favorable safety signal in multiple doses tested, including 2.88 mg/kg.
- Clinical safety, efficacy, and PK data are presented at ESMO posters 1804P and 1828P.

Corresponding author: Shawn Zhang; **Email:** Shawn.Zhang@ambrx.com

Abbreviations: PSMA, prostate-specific membrane antigen; **PEG**, poly-ethylene glycol; **PK**, pharmacokinetics; **FIH**, first-in-human; **MMAE**, monomethyl auristatin E; **PDX**, patient-derived xenograft; **CDX**, cell line-derived xenograft; **LLOQ**, lower limit of quantitation; **Cyno**, cynomolgus monkey; **NHP**, non-human primate; **HNSTD**, highest non-severely toxic dose.

Sponsored by Ambrx, Inc.

RESULTS

Figure 1. In Vitro Cytotoxic Activity in Tumor Cells

ARX517 displays selective in vitro cytotoxicity in PSMA-expressing tumor cell line cultures

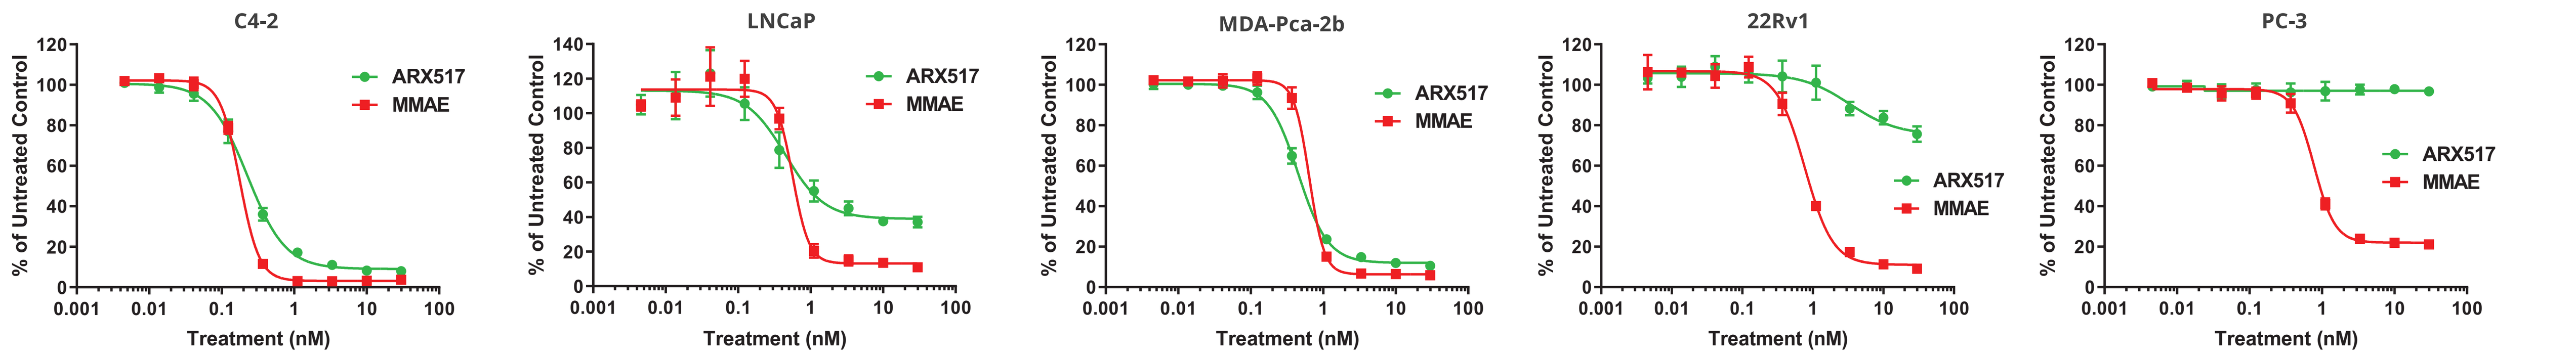
C4-2
LNCaP
MDA-Pca-2b
22Rv1
PC-3
ARX517
MMAE
% of Untreated Control
Treatment (nM)

Cell Line	PSMA Surface Number (QiFiKit)	ARX517 IC_{50} (nM)	ARX517 E_{max} (%)	MMAE IC_{50} (nM)	MMAE E_{max} (%)
C4-2	87,000	0.228	92	0.178	97
LNCaP	136,000	0.525	63	0.640	89
MDA-PCa-2b	46,000	0.451	89	0.653	94
22Rv1	6,000	>30	≥25	0.802	91
PC-3	<1000	>30	0	0.778	79

Figure 2. Efficacy in Enzalutamide-Sensitive and –Resistant Tumor Models, A. TM00298 PDX, B. C4-2 CDX

A

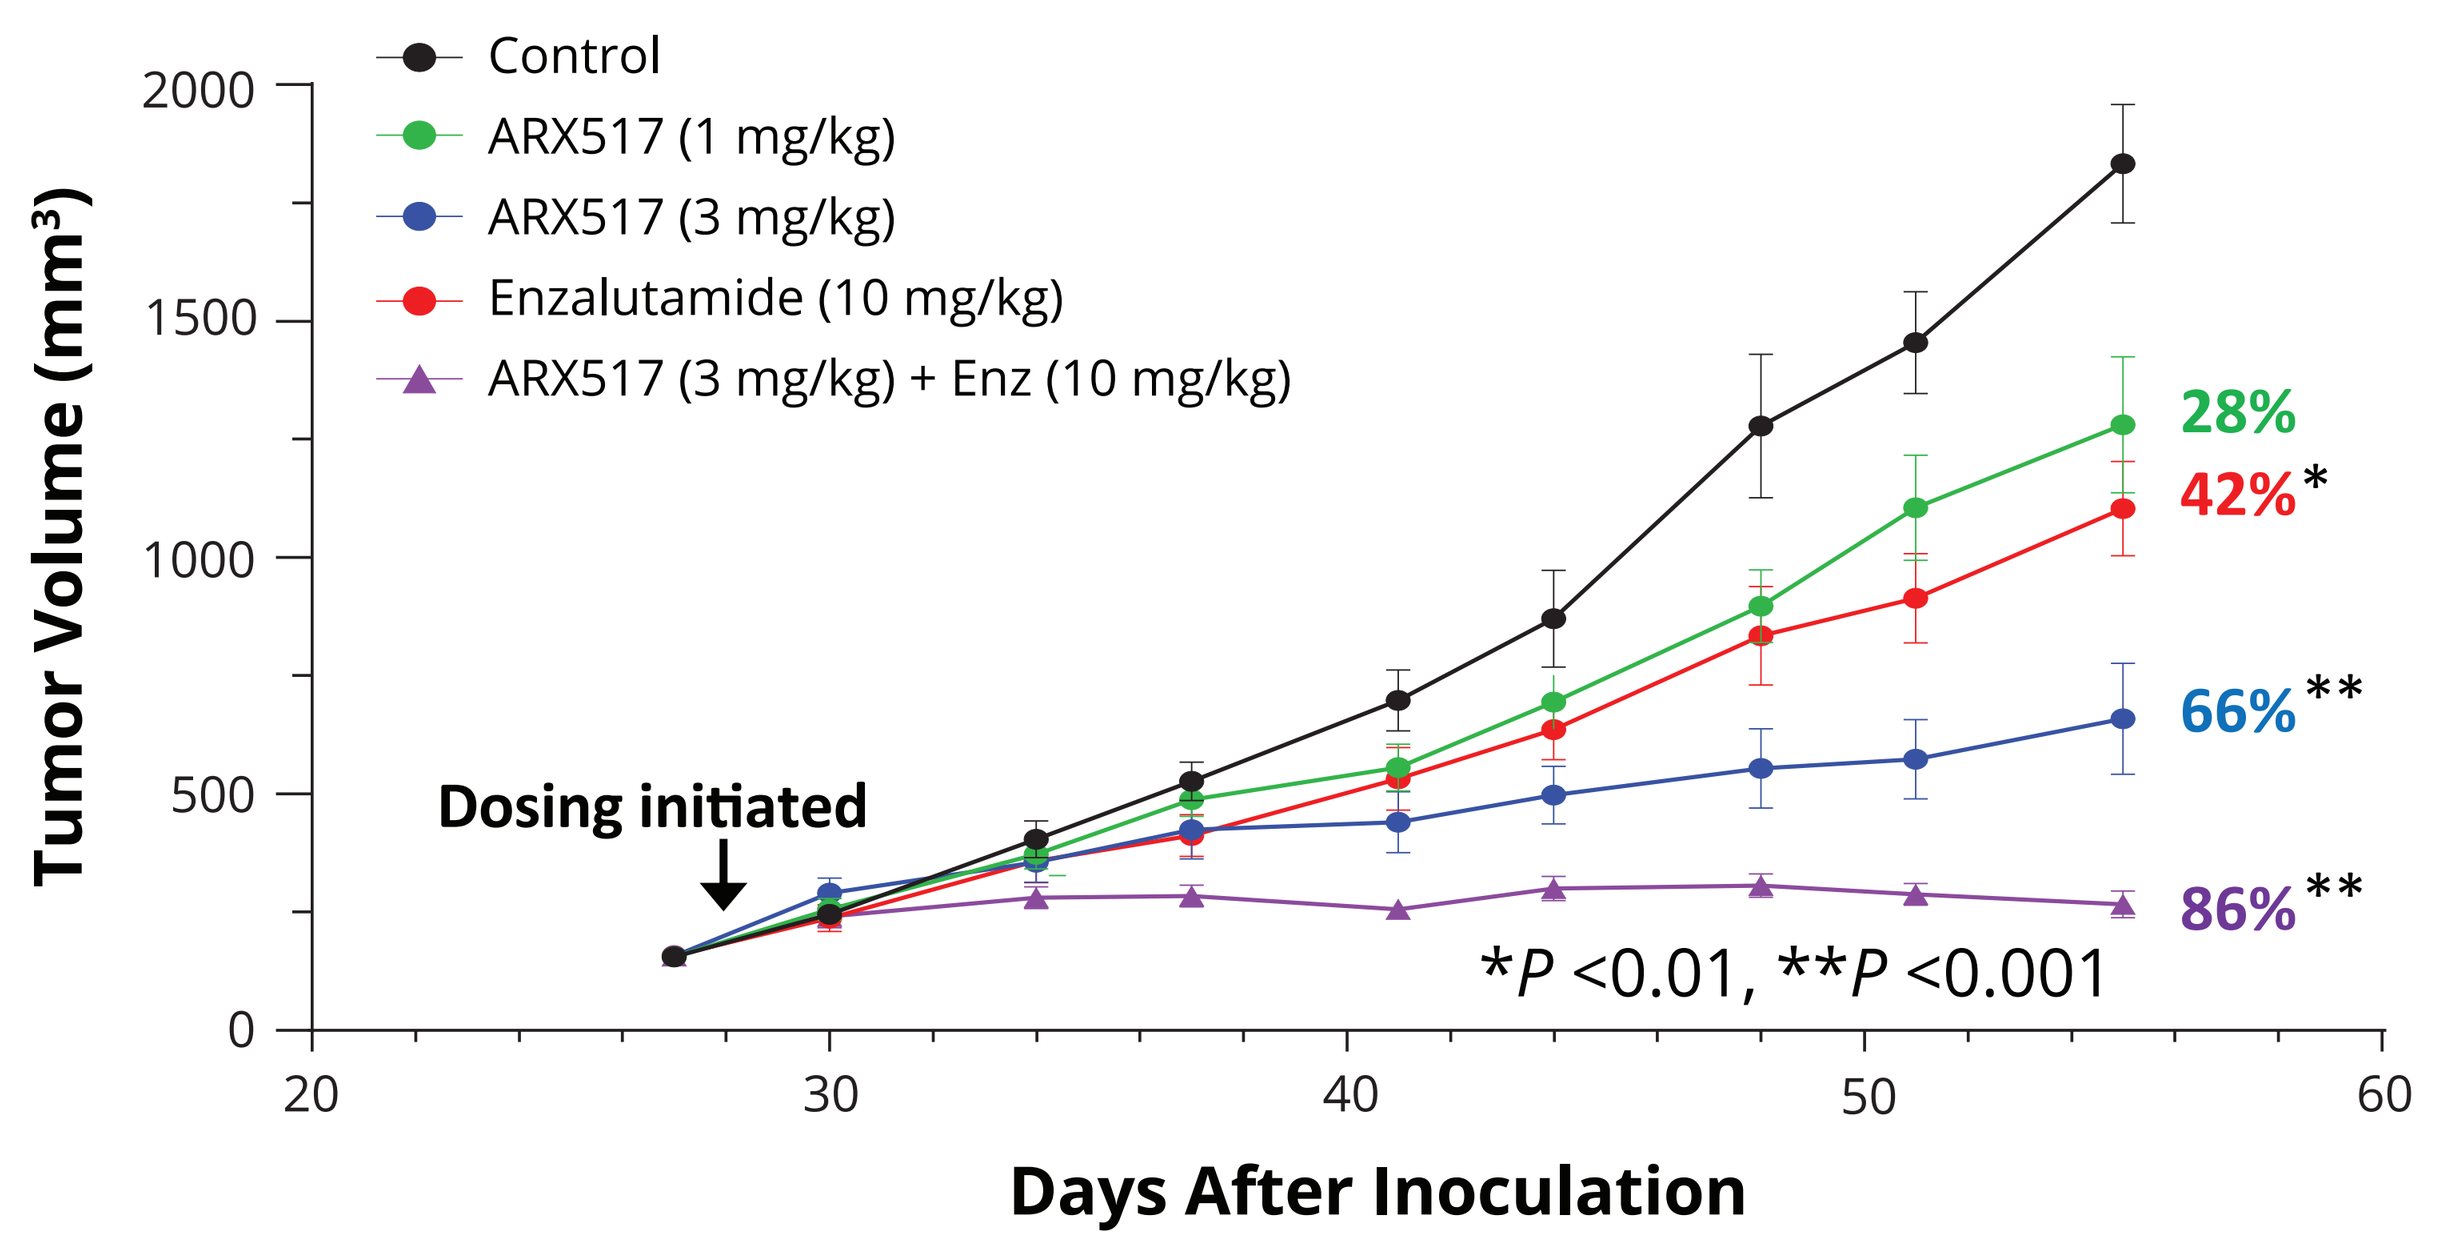
Control
ARX517 (1 mg/kg)
ARX517 (3 mg/kg)
Enzalutamide (10 mg/kg)
ARX517 (3 mg/kg) + Enz (10 mg/kg)
Tumor Volume (mm³)
Dosing initiated
28%
42%*
66%**
86%**
*P <0.01, **P <0.001
Days After Inoculation

NCG mice were subcutaneously implanted with enzalutamide-sensitive TM00298 patient-derived tumor cells. When tumors reached 100-200 mm³, mice were dosed with the indicated test articles; ARX517 was dosed i.v. QWx4 and enzalutamide was administered p.o. QDx18. Graph shows mean tumor volumes ± SEM over time.

B

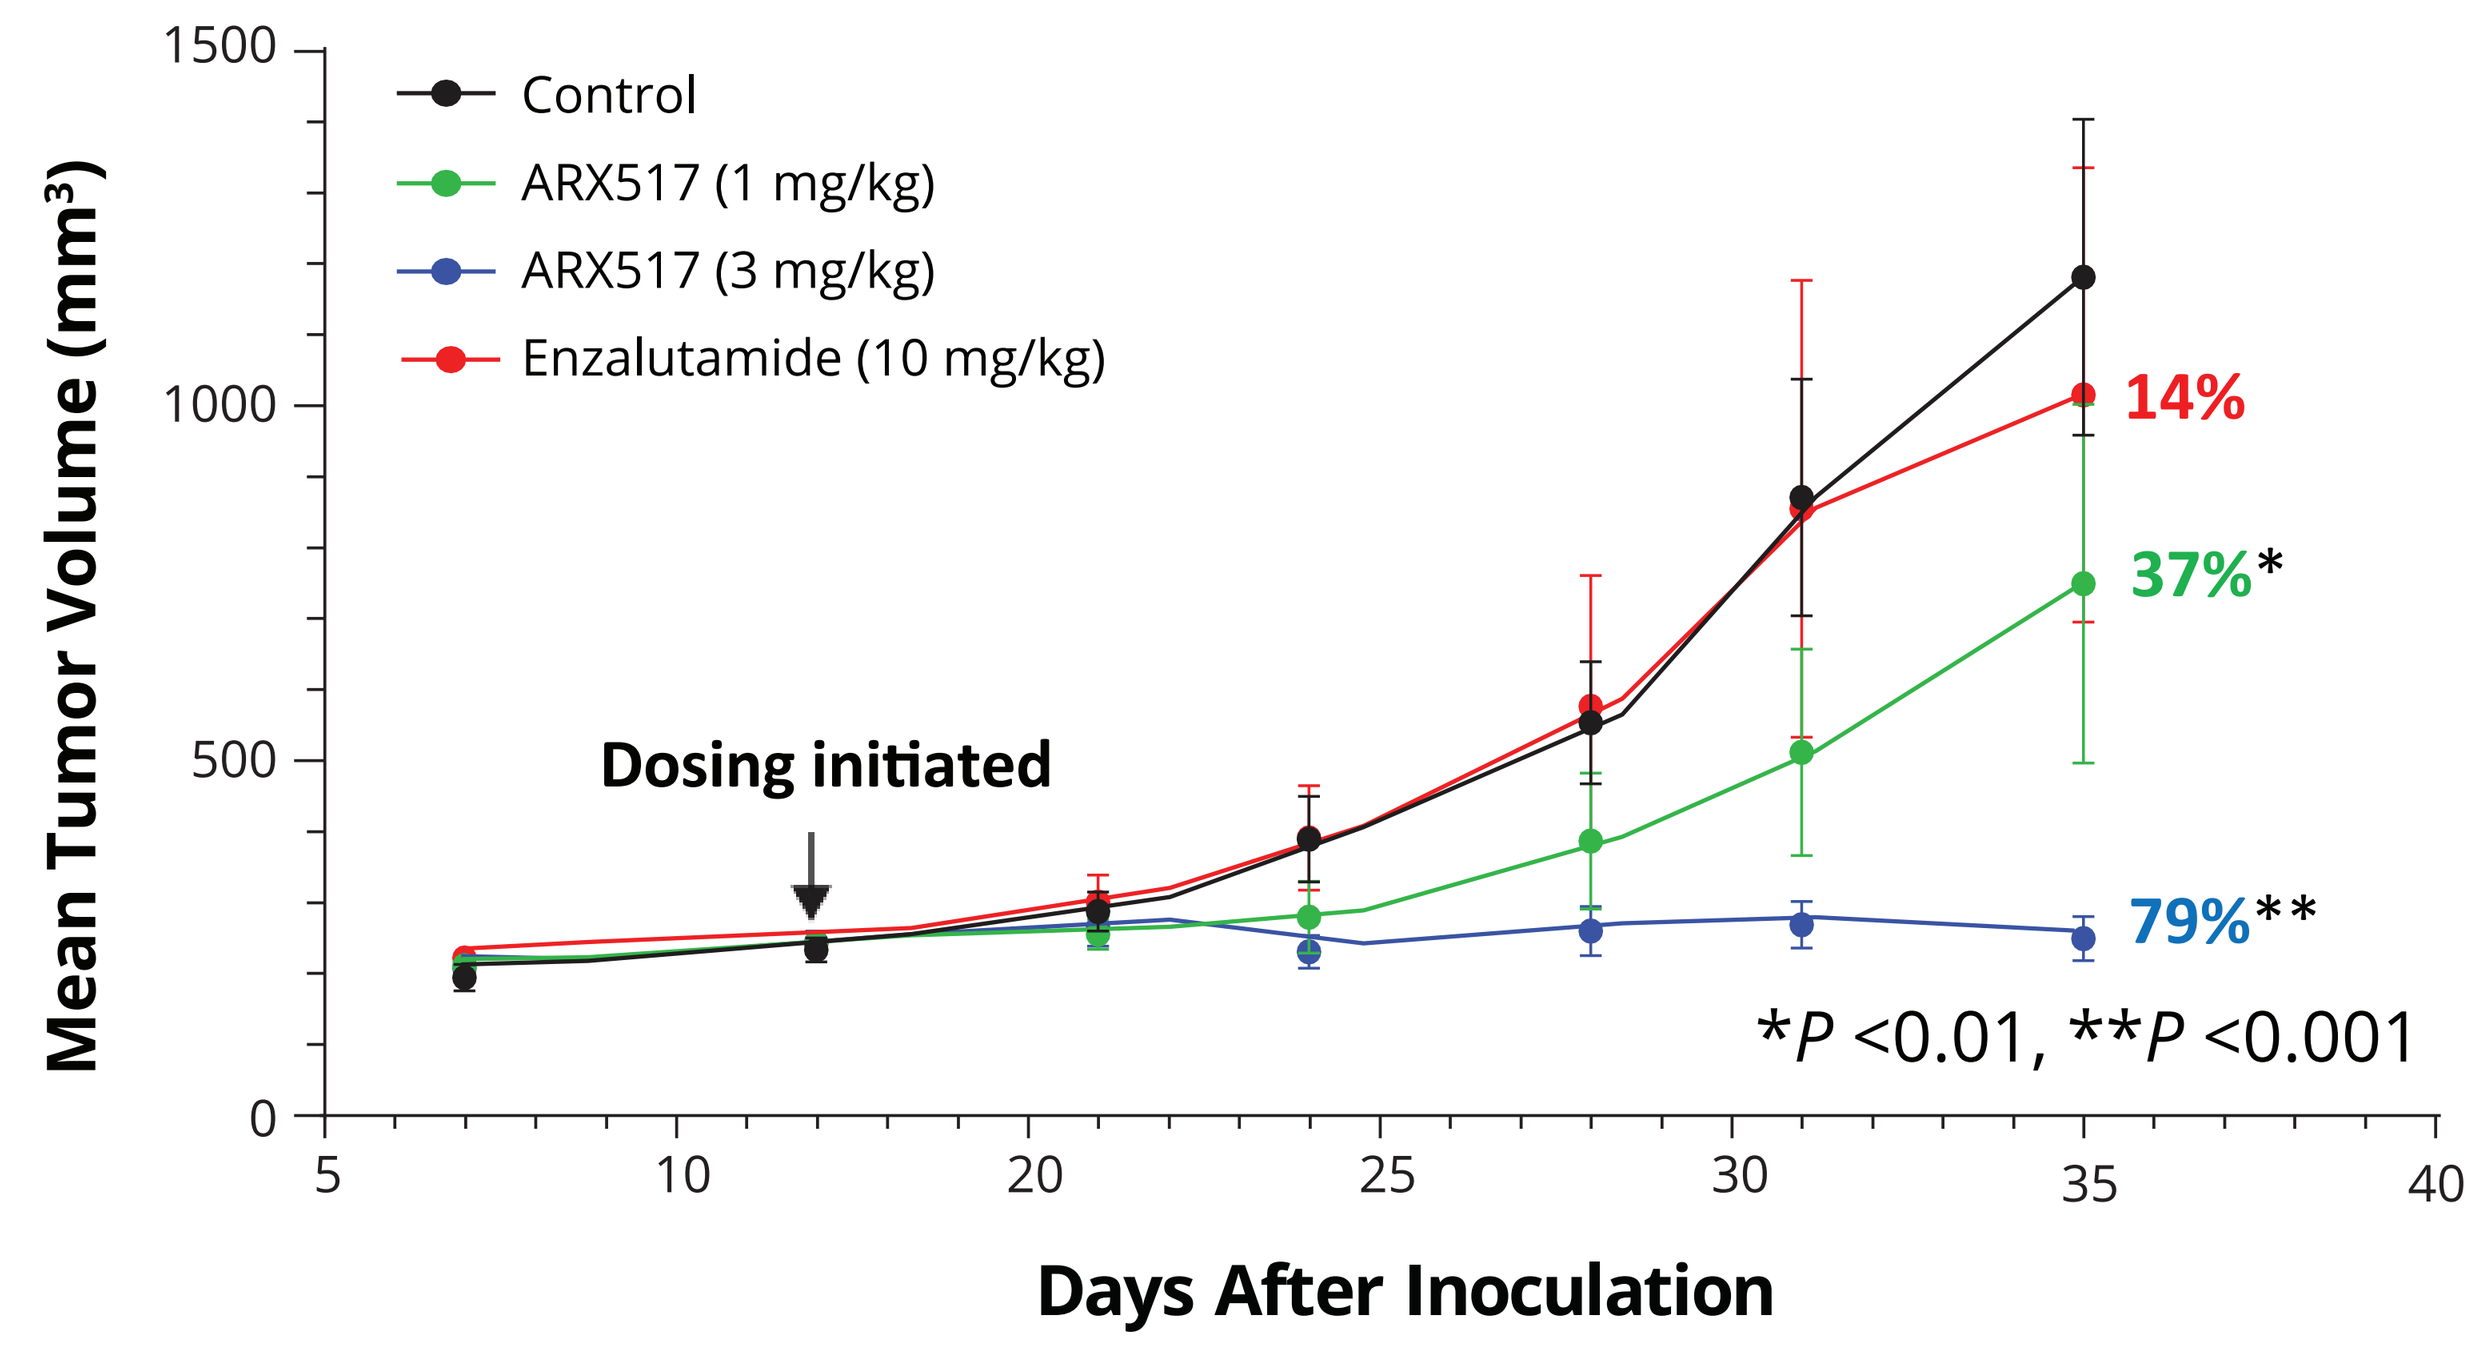
Control
ARX517 (1 mg/kg)
ARX517 (3 mg/kg)
Enzalutamide (10 mg/kg)
Mean Tumor Volume (mm³)
Dosing initiated
14%
37%*
79%**
*P <0.01, **P <0.001
Days After Inoculation

In an enzalutamide-resistant C4-2 prostate CDX model, 3 weekly doses of ≥1 mg/kg ARX517 (QWx3, IV) significantly inhibited tumor growth in a dose-dependent manner, whereas daily 10 mg/kg enzalutamide (QDx21, PO) treatment was ineffective.

Figure 3. Stability in Circulation

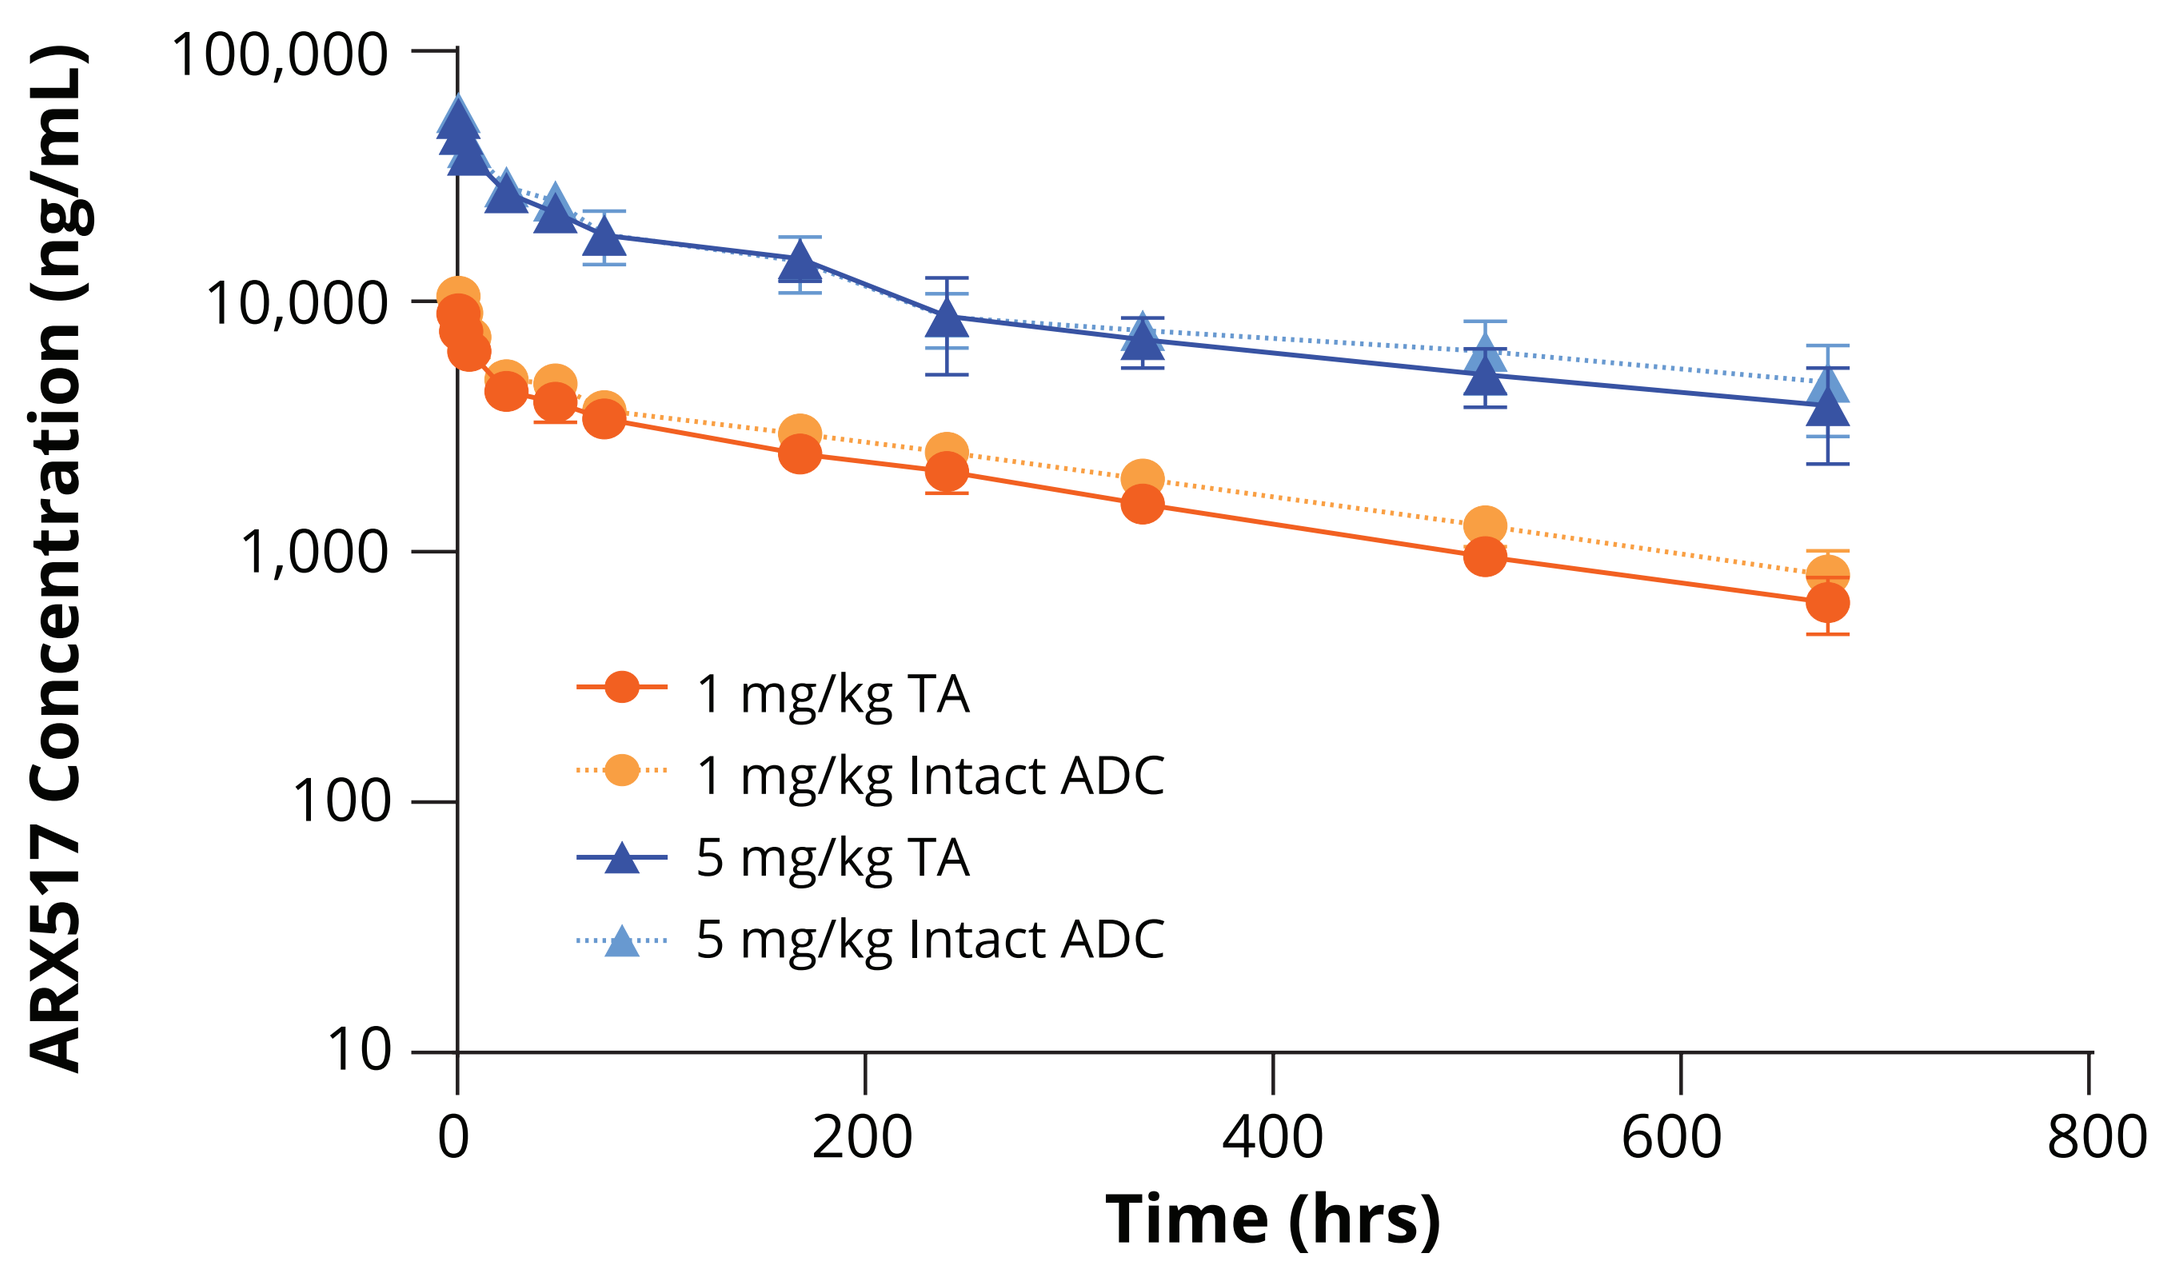
ARX517 Concentration (ng/mL)
1 mg/kg TA
1 mg/kg Intact ADC
5 mg/kg TA
5 mg/kg Intact ADC
Time (hrs)

Mice were dosed once intravenously with 1 mg/kg or 5 mg/kg ARX517, and samples were measured in Total Antibody (TA) and Intact ADC PK methods. The Intact ADC method only detects DAR2 ADCs which allows detection of any drug-linker loss when compared to TA curves. TA and Intact ADC curves were completely overlapped through study end, confirming the high stability of ARX517 in circulation with an extended half-life of 11 or 15 days.

Figure 4. Minimal Payload Release in Monkey Tox Study

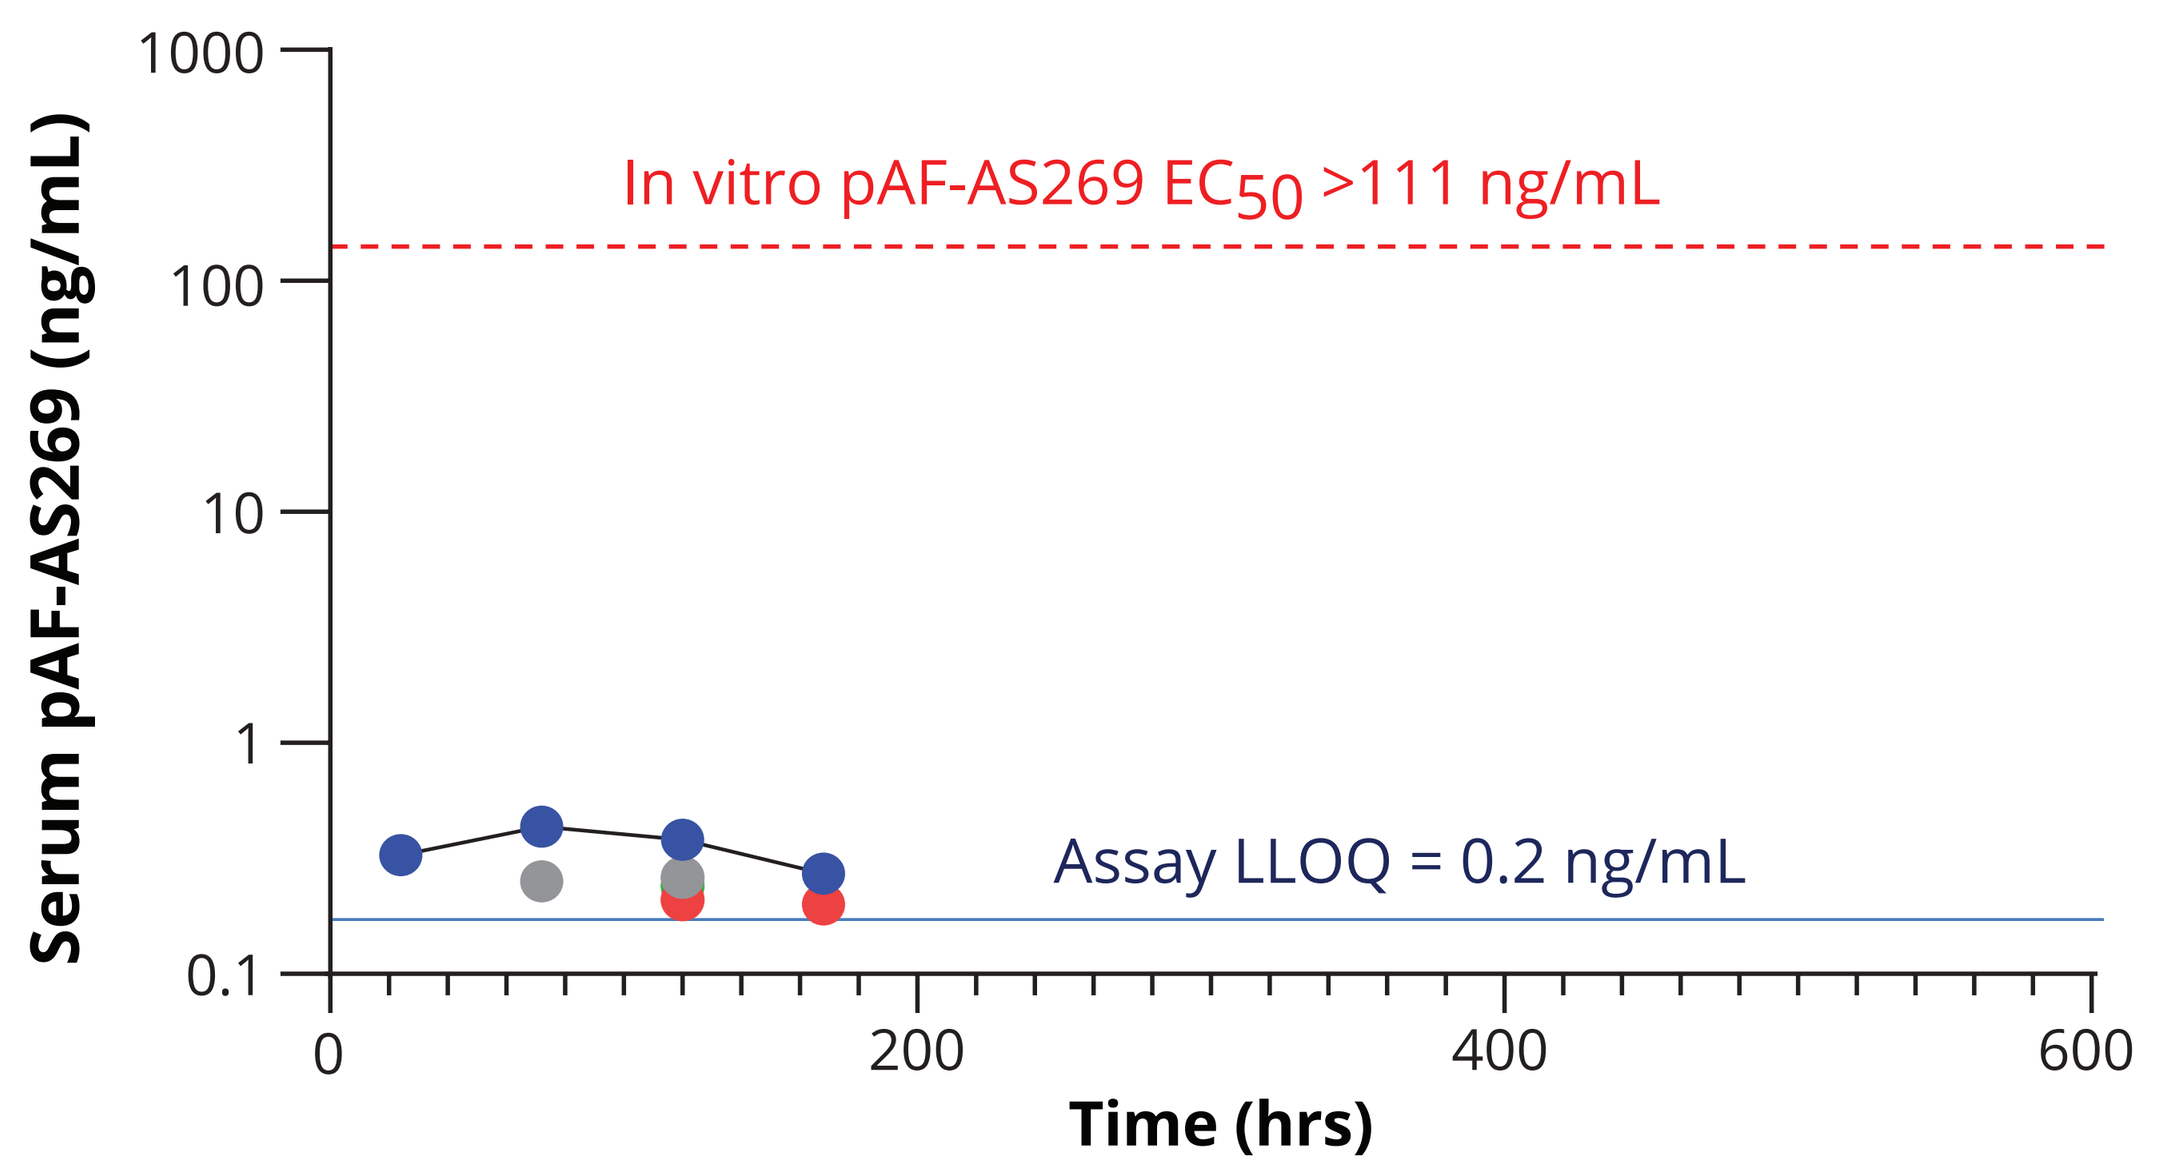
Serum pAF-AS269 (ng/mL)
In vitro pAF-AS269 EC50 >111 ng/mL
Assay LLOQ = 0.2 ng/mL
Time (hrs)

In the repeat-dose cynomolgus study, the payload pAF-AS269 slowly appeared in circulation at very low levels, just above the assay limit of quantitation (0.2 ng/mL). The serum pAF-AS269 levels were much lower than the pAF-AS269 IC_{50} value (>100 nM=111 ng/mL) observed in in vitro assays.

Figure 5. Monkey Toxicology & Therapeutic Index

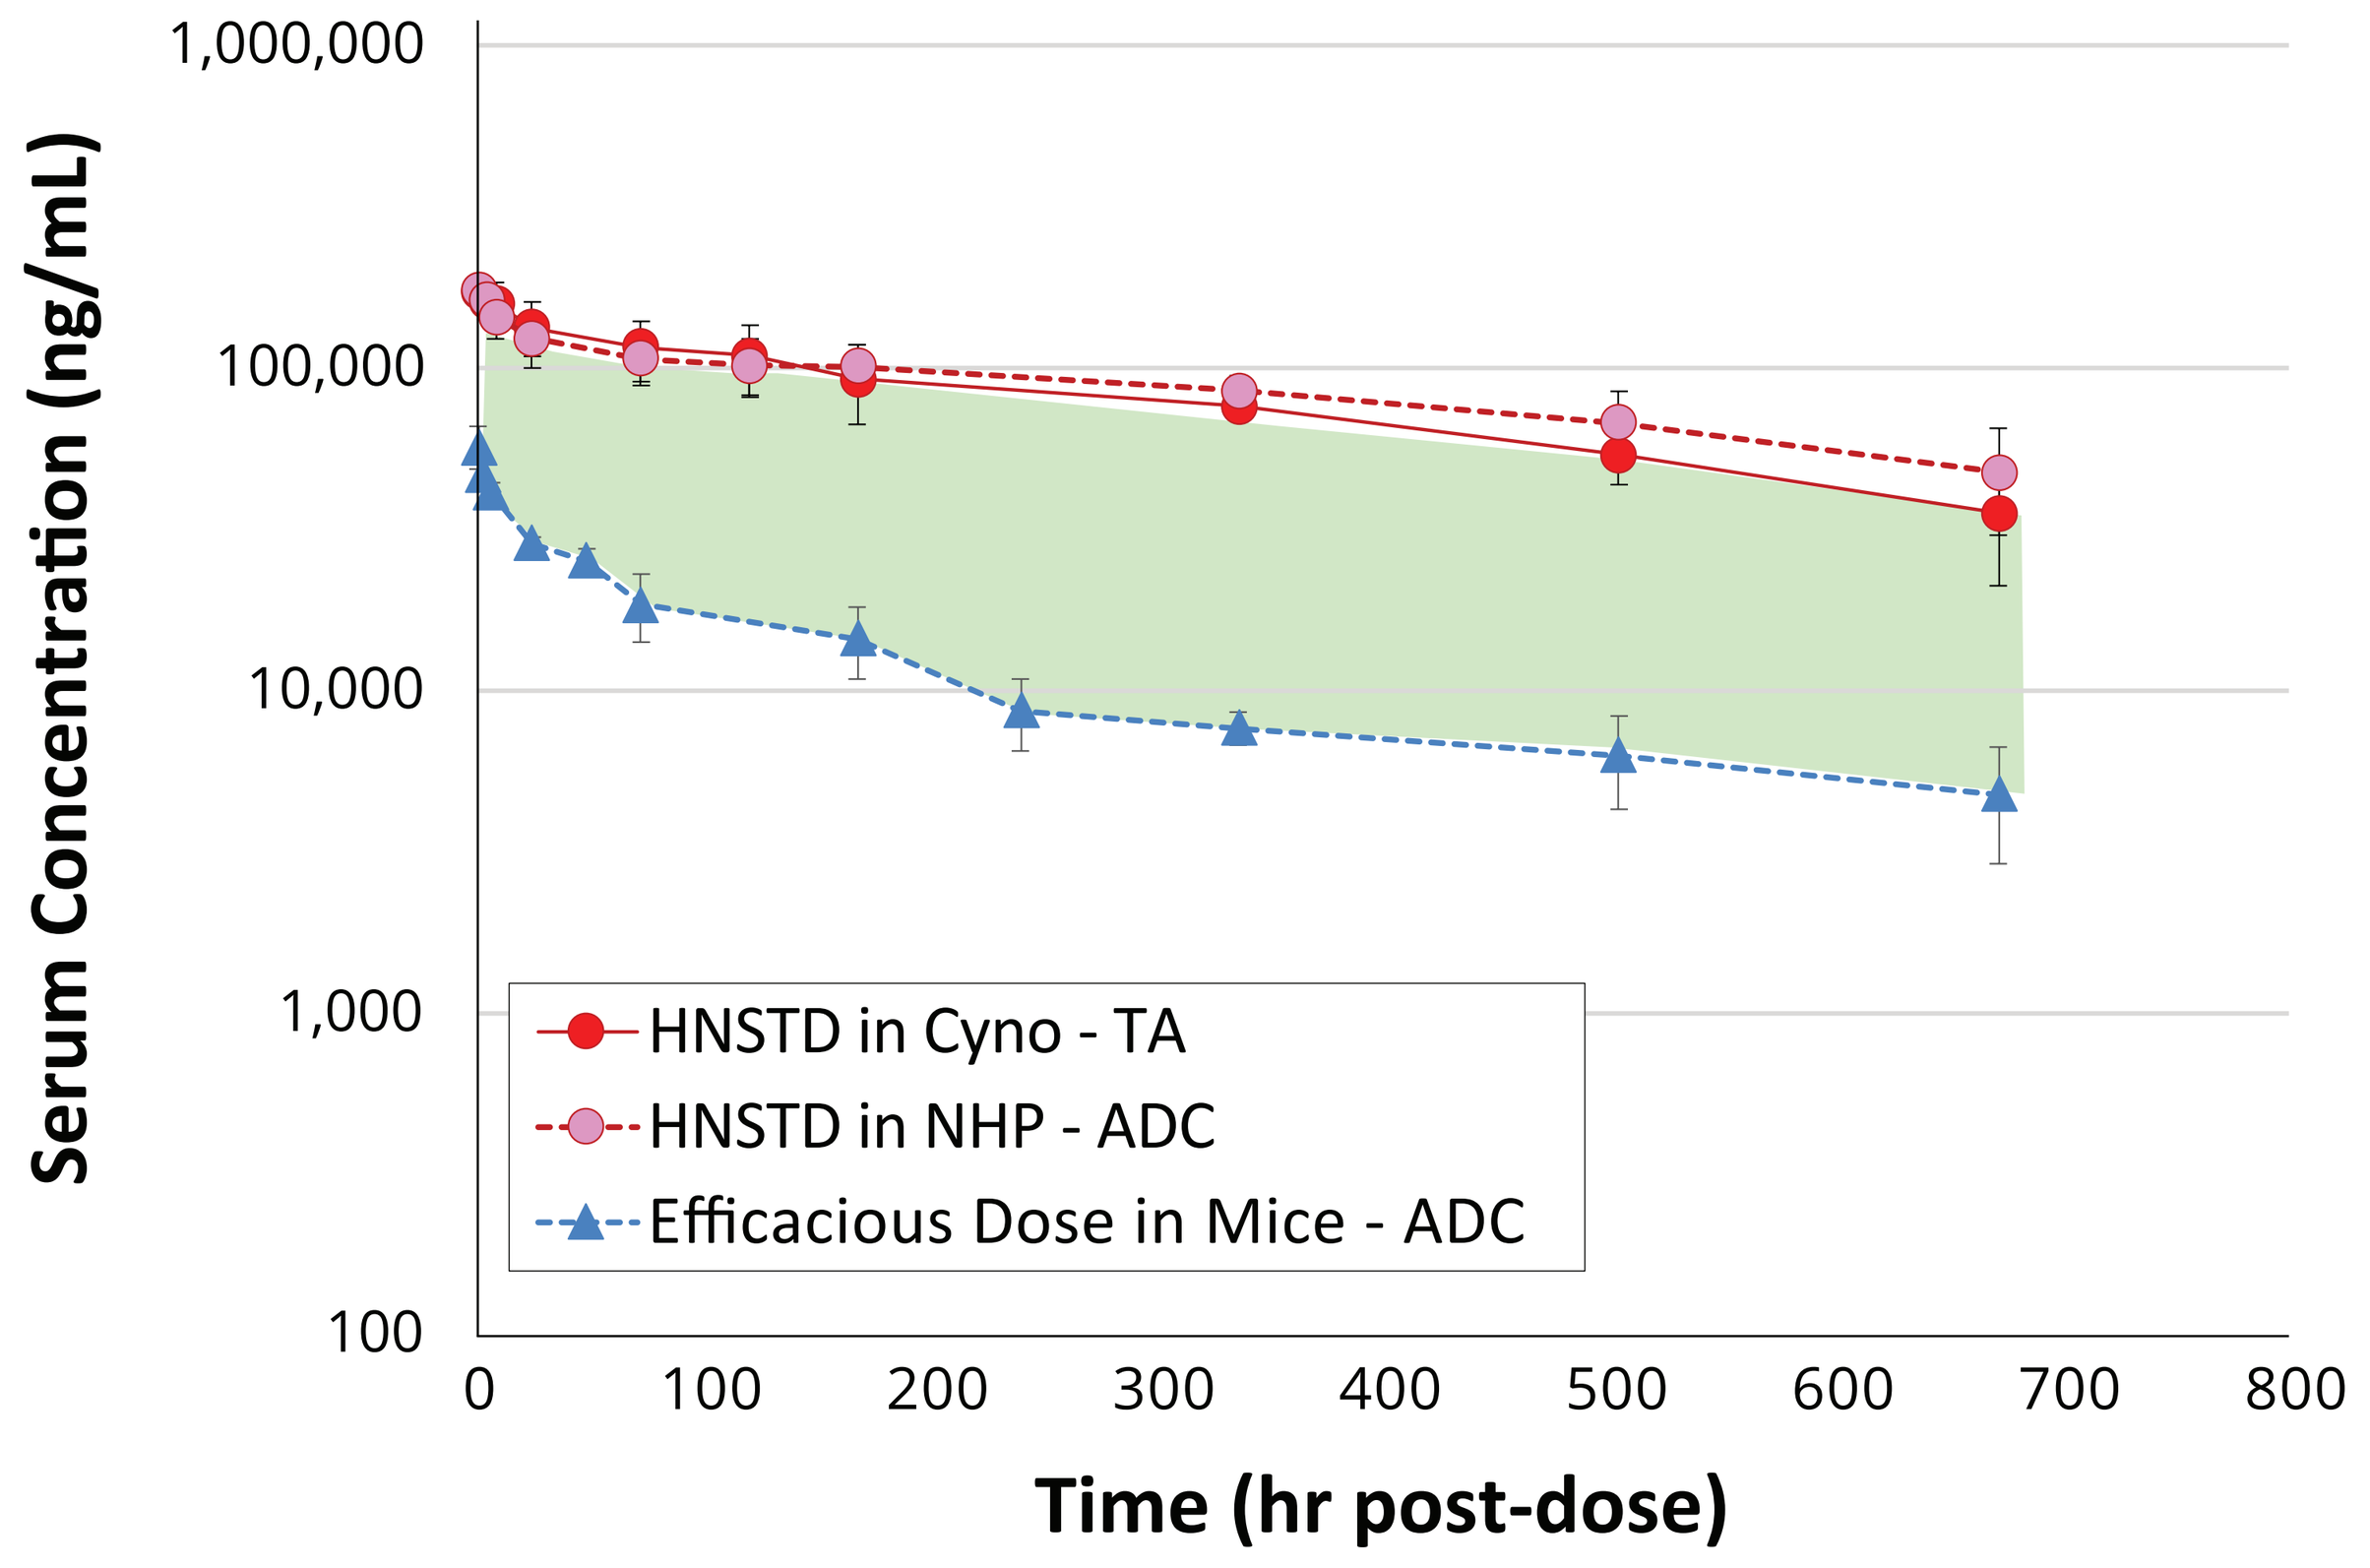
Serum Concentration (ng/mL)
HNSTD in Cyno - TA
HNSTD in NHP - ADC
Efficacious Dose in Mice - ADC
Time (hr post-dose)

ARX517 (cynomolgus cross-reactive) was administered to male cynomolgus monkeys once every 3 weeks for a total of 2 doses with a 6-week recovery period. Histopathologic findings were primarily observed in the kidney and liver. At the HNSTD, ARX517 serum exposure was greater than ARX517 exposure at a pharmacologically active dose in mice, demonstrating a clear therapeutic index.

CONCLUSION

- **ARX517 exhibits anti-tumor activity in preclinical enzalutamide-sensitive and –resistant prostate cancer models, with high stability in circulation.**
- **ARX517 demonstrates a clear therapeutic index in IND-enabling studies.**
- **The upregulation of PSMA by enzalutamide in mCRPC patients and our preclinical combination data provide scientific rationale to investigate the potential benefit of ARX517 and enzalutamide combination therapy in mCRPC patients.**
- **ARX517 is currently being evaluated in a multi-center Phase 1 clinical trial in the US (ARX517-2011, APEX-01, [NCT04662580])**

References

1. Linder S, et al. *Endocrine-Related Cancer* 2019; 26: R31-R52.
2. Staniszewski M, et al. *Int J Mol Sci* 2021; 22, 7431.
3. Rosar F, et al. *Cancers* 2022; 14(7): 1696.
4. Wondergem M, et al. *Clin Nucl Med* 2019; 44(7): 582-589

Disclosure

Shawn Zhang, Ph.D., is an employee of Ambrx, Inc.

Please see ESMO posters 1804P and 1828P for APEX-01 results

Copies of this poster obtained through QR (Quick Response) are for personal use only and may not be reproduced without written permission of the authors.

ESMO Poster 1804P ESMO Poster 1828P ESMO Poster 41P

PRESENTED AT EUROPEAN SOCIETY FOR MEDICAL ONCOLOGY (ESMO) ANNUAL MEETING, 20-24 OCTOBER 2023, MADRID, SPAIN