Phase 1 Dose Escalation Study of ARX788, a Next-Generation Anti-HER2 Antibody Drug Conjugate, in Heavily Pretreated Breast Cancer Patients

P01-04-01

Sophia Frentzas[1], Haeseong Park[2], George Thomas Budd[3], Vinod Ganju[4], Catherine Shannon[5], Sara A. Hurvitz[6], Katharine Cuff[7], Peter Lau[8], Richard Eek[9], Joyce O'Shaughnessy[10], Fran Boyle[11], Sandra Aung[12], Colin Hessel[12], Doug Greene[12], Bridget O'Keeffe[12], Jin Zhang[12], Quan Hong[12], Janice Lu[13]

[1]Monash Health, Melbourne, Australia; [2]Dana Farber Cancer Institute, Boston, MA; [3]Cleveland Clinic, Cleveland, OH; [4]Peninsula and South Eastern Haematology and Oncology Group, Australia; [5]Mater Cancer Care Centre, South Brisbane, Australia; [6]Fred Hutchinson Cancer Center, Seattle, WA; [7]Princess Alexandra Hospital, Woolloongabba, Australia; [8]Linear Clinical Research, Harry Perkins Institute for Medical Research, University of Western Australia, Nedlands, Australia; [9]Border Medical Oncology and Hematology, Albury, Australia; [10]Baylor University Medical Center, Texas Oncology and The US Oncology Network, Dallas, TX; [11]University of Sydney and Mater Hospital, Sydney, Australia; [12]Ambrx, Inc; [13]Feinberg School of Medicine, Northwestern University, Chicago, IL

BACKGROUND

- ARX788 is a next-generation HER2-targeted antibody-drug conjugate (ADC) with a highly stable conjugation to amberstatin-269 (AS269), a potent cytotoxic tubulin inhibitor (**Figure 1**). A proprietary site-specific oxime conjugation chemistry results in nearly identical pharmacokinetic (PK) profiles for the total antibody and ADC, and minimized systemic exposure to the free payload (**Figure 2**).[1,2]
- Breast cancer treatment has recently been transformed by ADCs, with the HER2-targeted ADC, fam-trastuzumab deruxtecan-nxki (T-DXd) providing survival benefits in both HER2-positive (HER2+) and HER2-low advanced breast cancer.[3,4]
- Clinical benefit of ARX788 in patients with HER2+ breast cancer has been reported for clinical trials conducted primarily in China.[1,5]
- The ACE-PanTumor-01 Phase 1 clinical trial in the US and Australia is one of two dose-finding studies for ARX788 in solid tumors that were completed with no DLT reported or MTD reached.[2]
- Here we present safety data and preliminary efficacy data in heavily pre-treated patients with HER2+ and HER2-low breast cancer treated with ARX788 monotherapy in the ACE-PanTumor-01 trial (ARX788-1711; NCT03255070).

Figure 1. ARX788 Construct

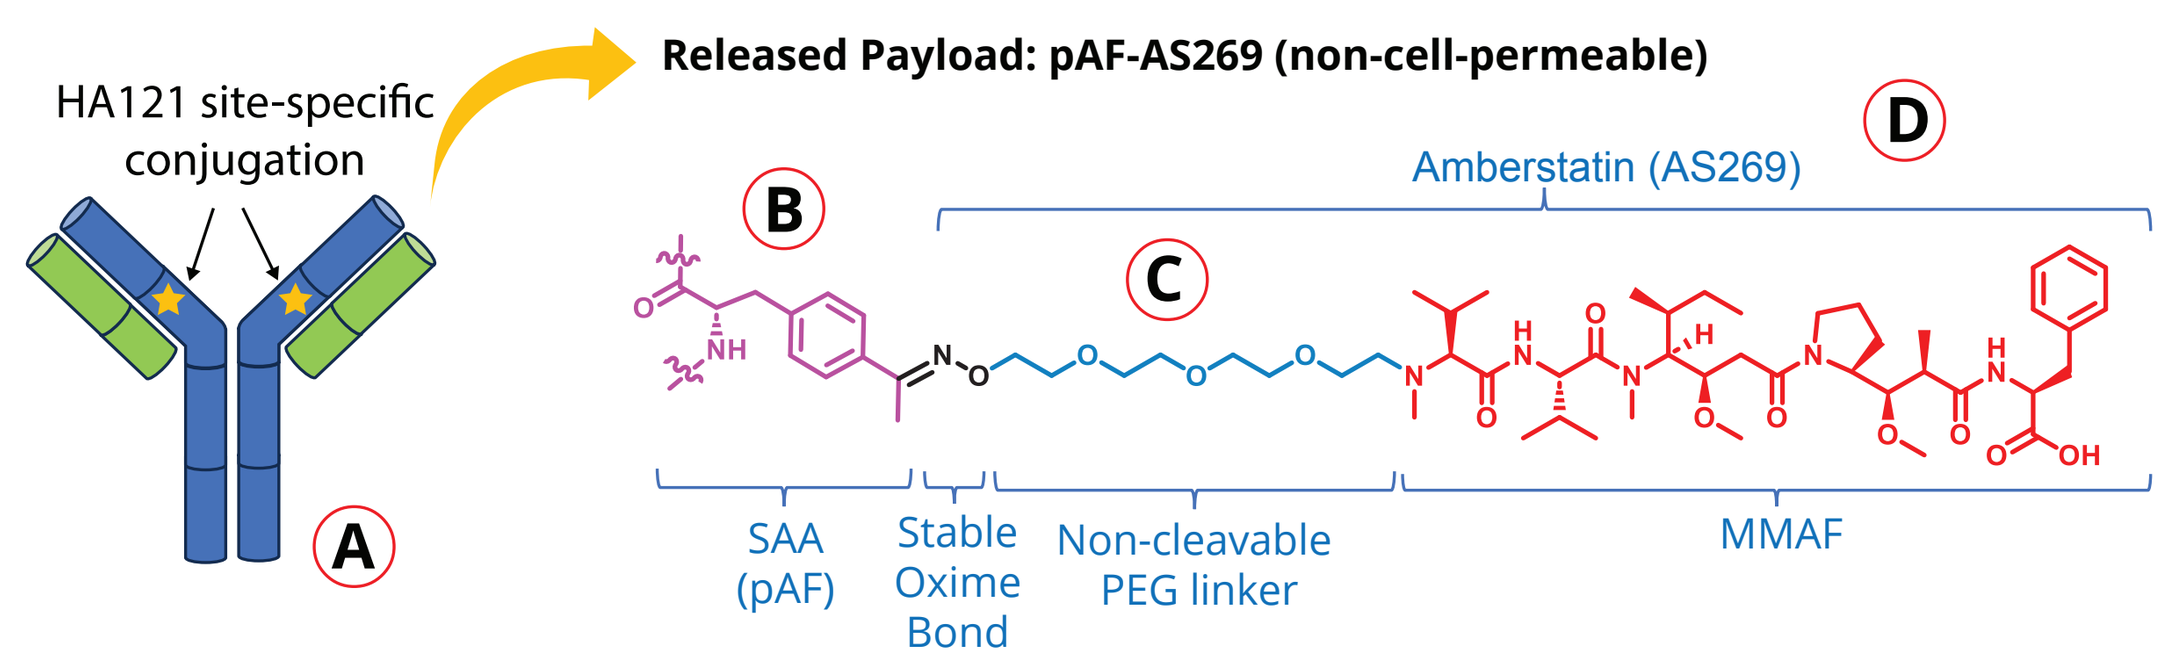
HA121 site-specific conjugation
Released Payload: pAF-AS269 (non-cell-permeable)
Amberstatin (AS269)
A
B
C
D
SAA (pAF)
Stable Oxime Bond
Non-cleavable PEG linker
MMAF

ARX788 is comprised of 4 key elements: (**A**) a humanized anti-HER2 antibody; (**B**) a payload covalently conjugated to a a synthetic amino acid (SAA), para-acetyl phenylalanine (pAF), genetically encoded and biosynthetically incorporated at amino acid position 121 on the heavy chain of the anti-HER2 antibody; (**C**) a non-cleavable PEG linker; and (**D**) a non-cell-permeable cytotoxic payload of a microtubule targeting antineoplastic agent (pAF-AS269).

Figure 2. Low Serum Concentration of ARX788 Payload

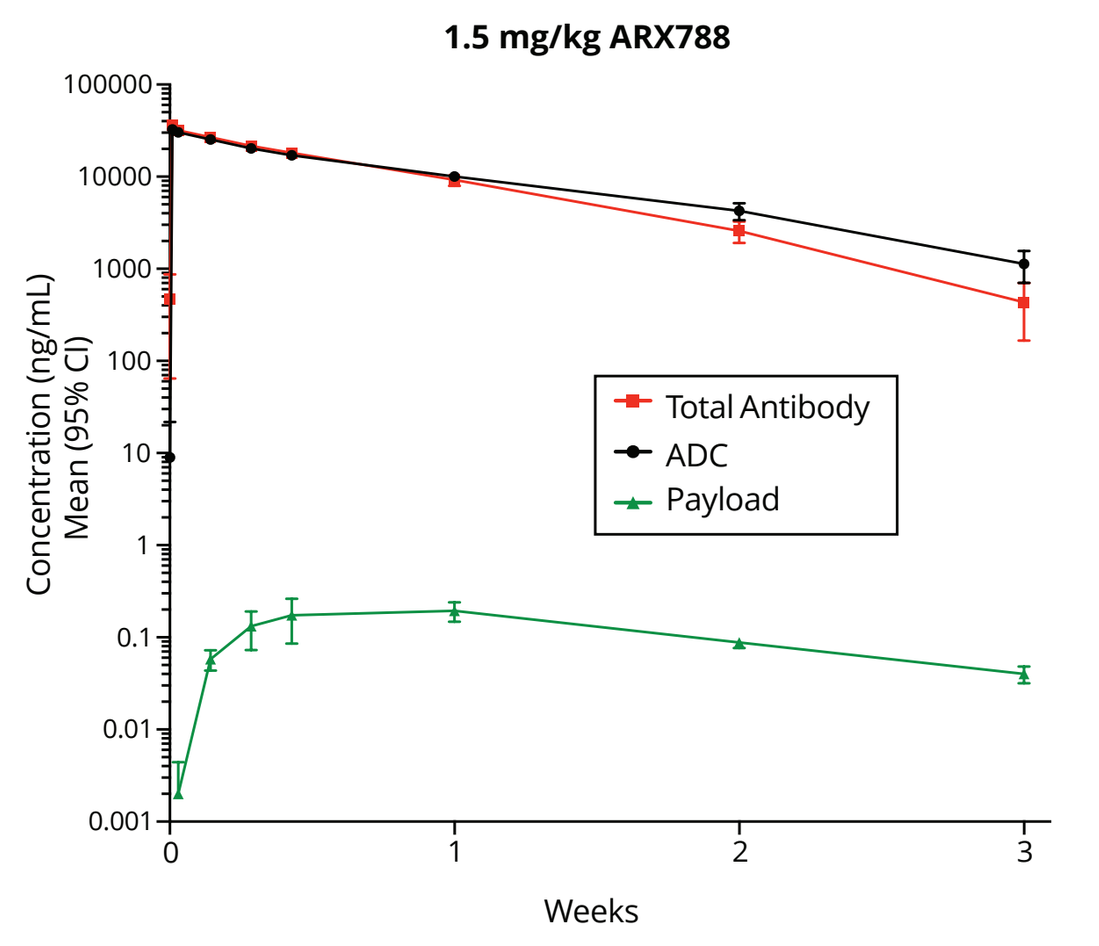
1.5 mg/kg ARX788
Concentration (ng/mL) Mean (95% CI)
100000
10000
1000
100
10
1
0.1
0.01
0.001
0
1
2
3
Weeks
Total Antibody
ADC
Payload

STUDY DESIGN

METHODS

- ARX788-1711 is a phase 1, multicenter, dose-escalation (Part 1) and expansion (Parts 2-3) study of ARX788 monotherapy in patients with advanced solid tumors with HER2 expression or mutation. Eligibility was open to patients for whom standard-of-care therapy had failed or was an unacceptable option.
- **Part 1** (dose escalation phase) explored doses of 0.66, 0.88, 1.1, 1.3, and 1.5 mg/kg every 3 weeks (Q3W) or every 4 weeks (Q4W). In the expansion phase, **Part 2** explored a frozen liquid formulation in additional dosing regimens, including 1.5 mg/kg Q6W and initial loading doses (1.7 mg/kg or 1.5 mg/kg) followed by reduced-level maintenance doses (1.5 mg/kg or 1.3 mg/kg, respectively) Q4W; and **Part 3** explored a lyophilized powder formulation at 1.6 mg/kg and 1.7 mg/kg Q3W. Patients were assessed for tumor response every 6 weeks (for Q3W and Q6W treatment schedules) or every 8 weeks (for Q4W treatment schedule). (**Figure 3**)
- Preliminary data for enrolled patients with HER2+ and HER2-low breast cancer treated at doses of 1.5, 1.6 and 1.7 mg/kg are presented (30 Oct 2023 data cutoff).

Figure 3. Study Design – (Part 1-3)

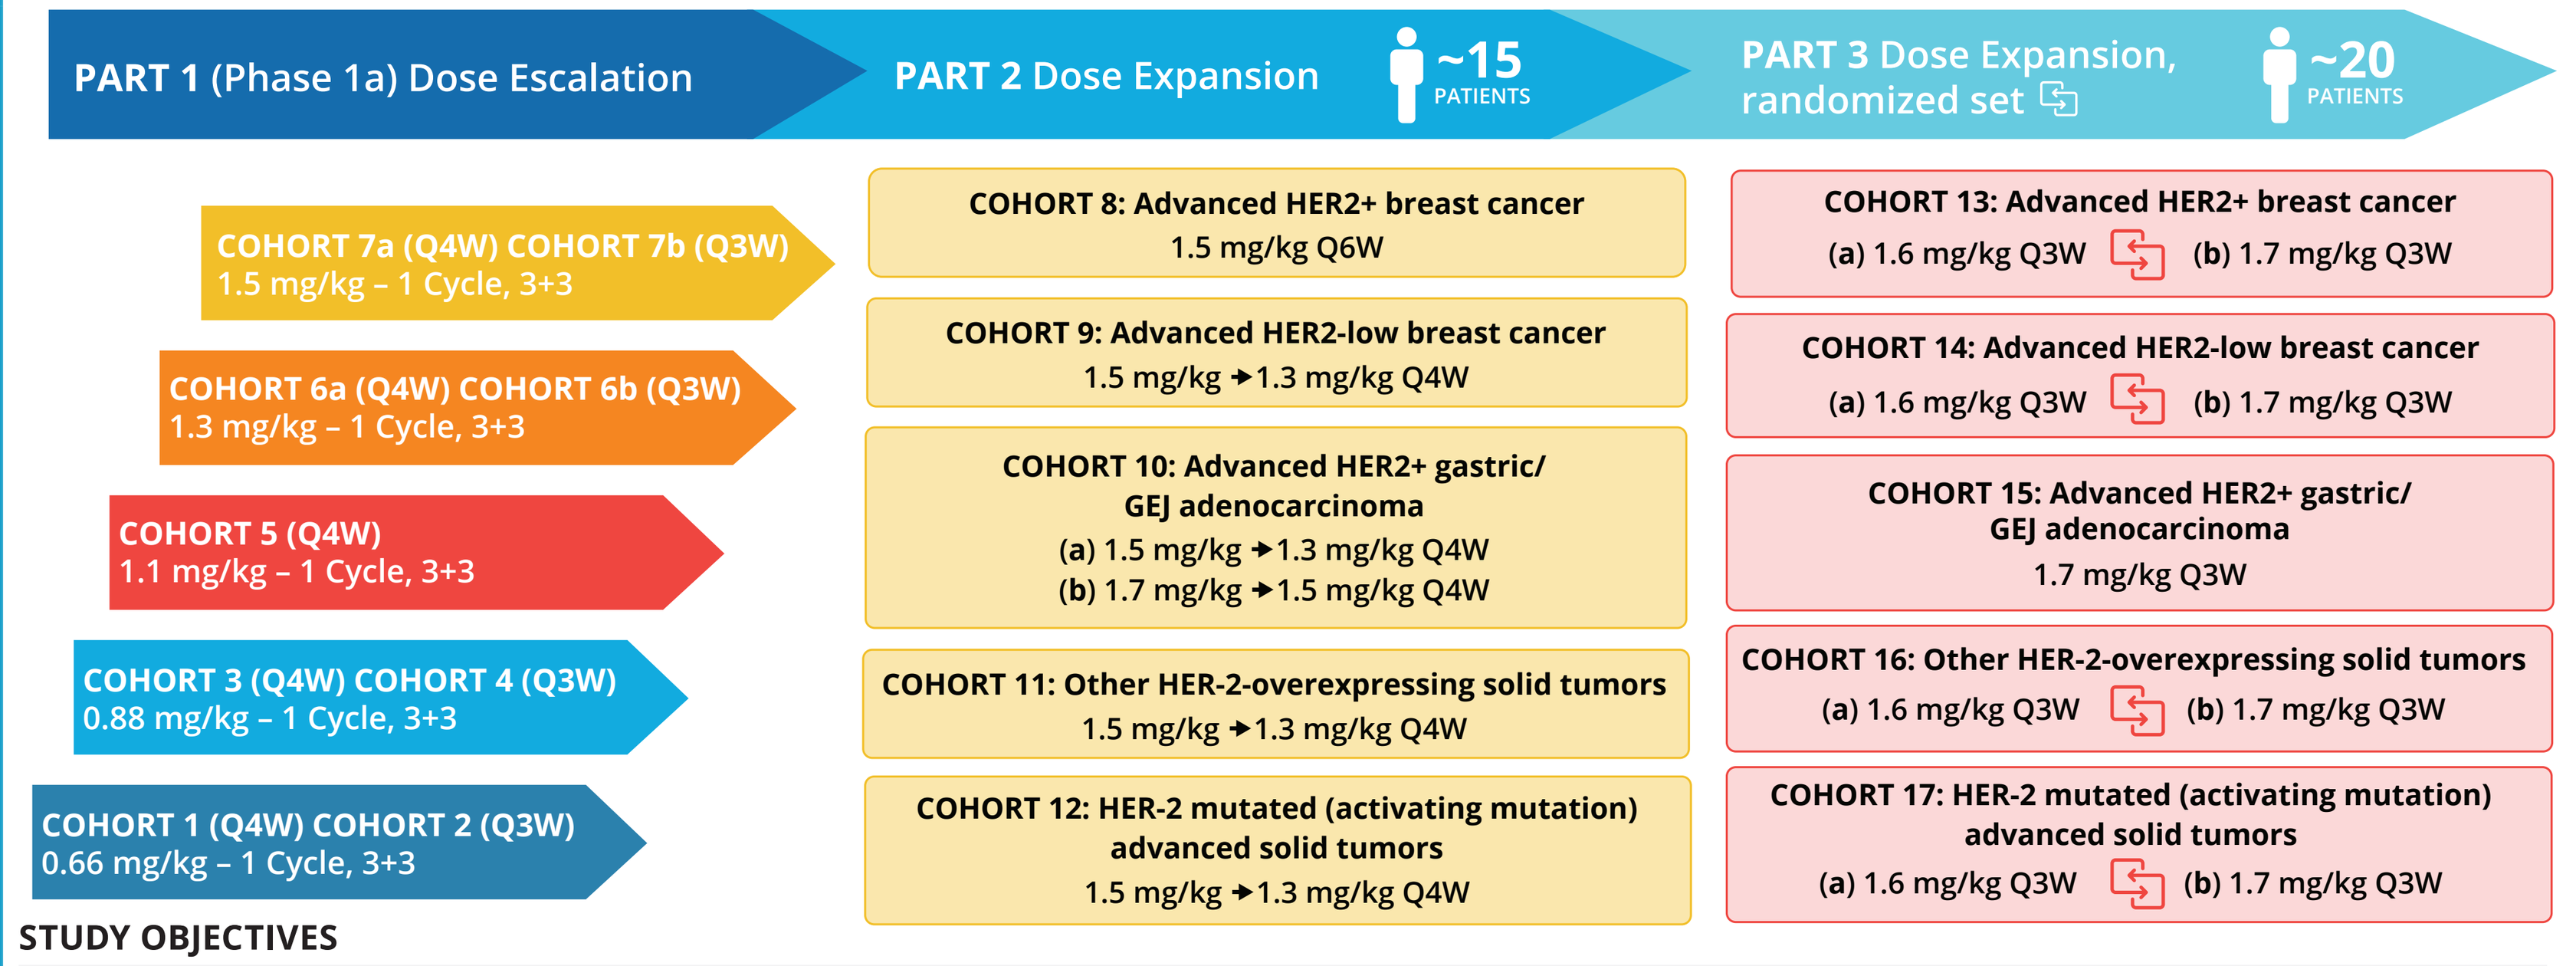
PART 1 (Phase 1a) Dose Escalation
PART 2 Dose Expansion ~15 PATIENTS
PART 3 Dose Expansion, randomized set ~20 PATIENTS
COHORT 7a (Q4W) COHORT 7b (Q3W) 1.5 mg/kg – 1 Cycle, 3+3
COHORT 6a (Q4W) COHORT 6b (Q3W) 1.3 mg/kg – 1 Cycle, 3+3
COHORT 5 (Q4W) 1.1 mg/kg – 1 Cycle, 3+3
COHORT 3 (Q4W) COHORT 4 (Q3W) 0.88 mg/kg – 1 Cycle, 3+3
COHORT 1 (Q4W) COHORT 2 (Q3W) 0.66 mg/kg – 1 Cycle, 3+3
COHORT 8: Advanced HER2+ breast cancer 1.5 mg/kg Q6W
COHORT 9: Advanced HER2-low breast cancer 1.5 mg/kg → 1.3 mg/kg Q4W
COHORT 10: Advanced HER2+ gastric/GEJ adenocarcinoma (a) 1.5 mg/kg → 1.3 mg/kg Q4W (b) 1.7 mg/kg → 1.5 mg/kg Q4W
COHORT 11: Other HER-2-overexpressing solid tumors 1.5 mg/kg → 1.3 mg/kg Q4W
COHORT 12: HER-2 mutated (activating mutation) advanced solid tumors 1.5 mg/kg → 1.3 mg/kg Q4W
COHORT 13: Advanced HER2+ breast cancer (a) 1.6 mg/kg Q3W (b) 1.7 mg/kg Q3W
COHORT 14: Advanced HER2-low breast cancer (a) 1.6 mg/kg Q3W (b) 1.7 mg/kg Q3W
COHORT 15: Advanced HER2+ gastric/GEJ adenocarcinoma 1.7 mg/kg Q3W
COHORT 16: Other HER-2-overexpressing solid tumors (a) 1.6 mg/kg Q3W (b) 1.7 mg/kg Q3W
COHORT 17: HER-2 mutated (activating mutation) advanced solid tumors (a) 1.6 mg/kg Q3W (b) 1.7 mg/kg Q3W

STUDY OBJECTIVES

- To determine safety and tolerability of ARX788
- To determine MTD and/or establish recommended phase 2 dose regimen
- Assessment of ORR based on RECIST v1.1

Number of subjects enrolled in each cohort: (**1**) n=3; (**2**) n=3; (**3**) n=7; (**4**) n=4; (**5**) n=3; (**6a**) n=4; (**6b**) n=8; (**7a**) n=3; (**7b**) n=0; (**8**) n=10; (**9**) n=20; (**10a**) n=6; (**10b**) n=0; (**11**) n=2; (**12**) n=1; (**13a**) n=3; (**13b**) n=3; (**14a**) n=6; (**14b**) n=6; (**15**) n=8; (**16a**) n=1; (**17a**) n=3; (**17b**) n=2. Subjects with HER2+ and HER2-low breast cancer were enrolled in cohorts 7-9 and 13a-14b.

RESULTS

Table 1. Baseline demographics & disease characteristics – breast cancer

Cohorts 7-9, 13, 14	HER2+ (N=11)	HER2 low (N=31)	All Breast Cancer (N=42)
Age, years – median (range)	54 (35, 67)	57 (41, 85)	56.5 (35, 85)
Female – n (%)	11 (100)	30 (97)	41 (98)
Race[a] – n (%)			
White	8 (73)	22 (71)	30 (71)
Asian	2 (18)	7 (23)	9 (21)
ECOG performance status – n (%)			
0	3 (27)	20 (65)	23 (55)
1	8 (73)	11 (35)	19 (45)
HR-positive – n (%)	9 (82)	24 (77)	33 (79)
HER2 status – n (%)			
IHC 1+	0 (0)	18 (58)	18 (43)
IHC 2+ (ISH negative)	0 (0)	13 (42)	13 (31)
IHC 2+ (ISH positive)	2 (18)	0 (0)	2 (5)
IHC 3+	8 (73)	0 (0)	8 (19)
IHC Unknown (ISH positive)	1 (9)	0 (0)	1 (2)
Sites of disease at enrollment – n (%)			
Lung	8 (73)	16 (52)	24 (57)
Liver	3 (27)	20 (65)	23 (55)
Bone	3 (27)	22 (71)	25 (60)
Prior advanced/metastatic treatment regimens			
Number – median (range)	6 (1, 9)	5 (1, 11)	5.5 (1, 11)
Advanced/metastatic treatment – n (%)			
Trastuzumab	10 (91)	4 (13)	14 (33)
Pertuzumab	6 (55)	3 (10)	9 (21)
Taxane	8 (73)	19 (61)	27 (64)
T-DM1	8 (73)	4 (13)	12 (29)
T-DXd	4 (36)	4 (13)	8 (19)
Tucatinib	3 (27)	3 (10)	6 (14)
CDK4/6 inhibitor	1 (9)	22 (71)	23 (55)
Capecitabine	7 (64)	22 (71)	29 (69)
Other chemotherapy	6 (55)	19 (61)	25 (60)
Endocrine	7 (64)	23 (74)	30 (71)

[a]One HER2+ and 2 HER2-low patients did not indicate race.

Table. 2. Adverse Event Summary

All	N=42
All Cause AEs, n (%)	
TEAEs: all Grade	41 (98)
Grade 3	15 (36)
Grade 4	0
Treatment-emergent SAEs	8 (19)
TEAEs leading to discontinuation	1 (2)
Fatal TEAEs (Grade 5)	0
Related to Study Drug, n (%)	
TRAEs: all Grade	39 (93)
Grade 3	10 (24)
Grade 4	0
Treatment-related SAEs	5 (12)
TEAEs leading to discontinuation	0
Fatal TEAEs (Grade 5)	0

Table 3. Most Frequent Grade 1-2 TRAEs (≥10%)

Preferred Term	N=42	Preferred Term	N=42
Dry eye	15 (36)	Nausea	6 (14)
Alopecia	15 (36)	Pneumonitis	6 (14)
Vision blurred	14 (33)	Photophobia	6 (14)
Fatigue	11 (26)	Decreased appetite	5 (12)
Dry mouth	8 (19)	Keratitis	5 (12)

Table 4. Summary of Grade 3 TRAEs; there were no Grade 4 or 5 TRAEs

Preferred Term	N=42	
	Grade 3	Grade 4/5
Patients with any TEAE	**10 (24)**	**0**
Aspartate Aminotransferase Increased	2 (5)	0
Blood Alkaline Phosphatase Increased	2 (5)	0
Gamma-Glutamyltransferase Increased	2 (5)	0
Nausea	2 (5)	0
Visual Impairment	2 (5)	0
Alanine Aminotransferase Increased	1 (2)	0
Blood Bilirubin Increased	1 (2)	0
Corneal Thinning	1 (2)	0
Decreased Appetite	1 (2)	0
Hypercalcemia	1 (2)	0
Hypokalemia	1 (2)	0
Organizing Pneumonia[a]	1 (2)	0
Pneumonia Parainfluenzae Viral	1 (2)	0
Pneumonitis[b]	1 (2)	0
Vomiting	1 (2)	0

[a]Patient with ongoing history of asthma and pulmonary metastases developed Grade 3 organizing pneumonia 357 days after first dose and 36 days after the last dose of ARX788. The event resolved within 9 days following treatment with antibiotics and dexamethasone (for brain metastases).

[b]Patient with ongoing history of cough developed Grade 3 pneumonitis 100 days after first and 16 days after last dose of ARX788. The event resolved within 42 days after treatment with antibiotics and prednisolone.

Table 5. TRAEs of Special Interest

All, N=42	Grade 1-2	Grade 3	Grade 4/5		Grade 1-2	Grade 3	Grade 4/5
Patients with any eye disorders, n (%)[a]	**27(64)**	**2(5)**	**0**	**Patients with any pneumonitis/ILD event, n (%)**	6(14)	2(5)	0
Dry eye	15(36)	0	0	Pneumonitis	6(14)	1 (2)[b]	0
Vision blurred	14(33)	0	0	Organizing pneumonia	0	1 (2)[c]	0
Photophobia	6(14)	0	0				
Keratitis	5(12)	0	0				
Visual impairment	2(5)	2(5)	0				

[a]Specific eye disorders listed for those occurring with a ≥10% frequency; eye disorders with any frequency included in the total.

[b]Grade 3 pneumonitis event occurred in a patient with ongoing history of cough 100 days after first and 16 days after the last dose of ARX788. The event resolved within 42 days after treatment with antibiotics and prednisolone.

[c]Patient with ongoing history of asthma and pulmonary metastases developed Grade 3 organizing pneumonia 357 days after first dose and 36 days after the last dose of ARX788. The event resolved within 9 days following treatment with antibiotics and dexamethasone (for brain metastases).

Figure 4. Anti-tumor Activity of ARX788 in Heavily Pretreated HER2+ and HER2-low Breast Cancer Patients

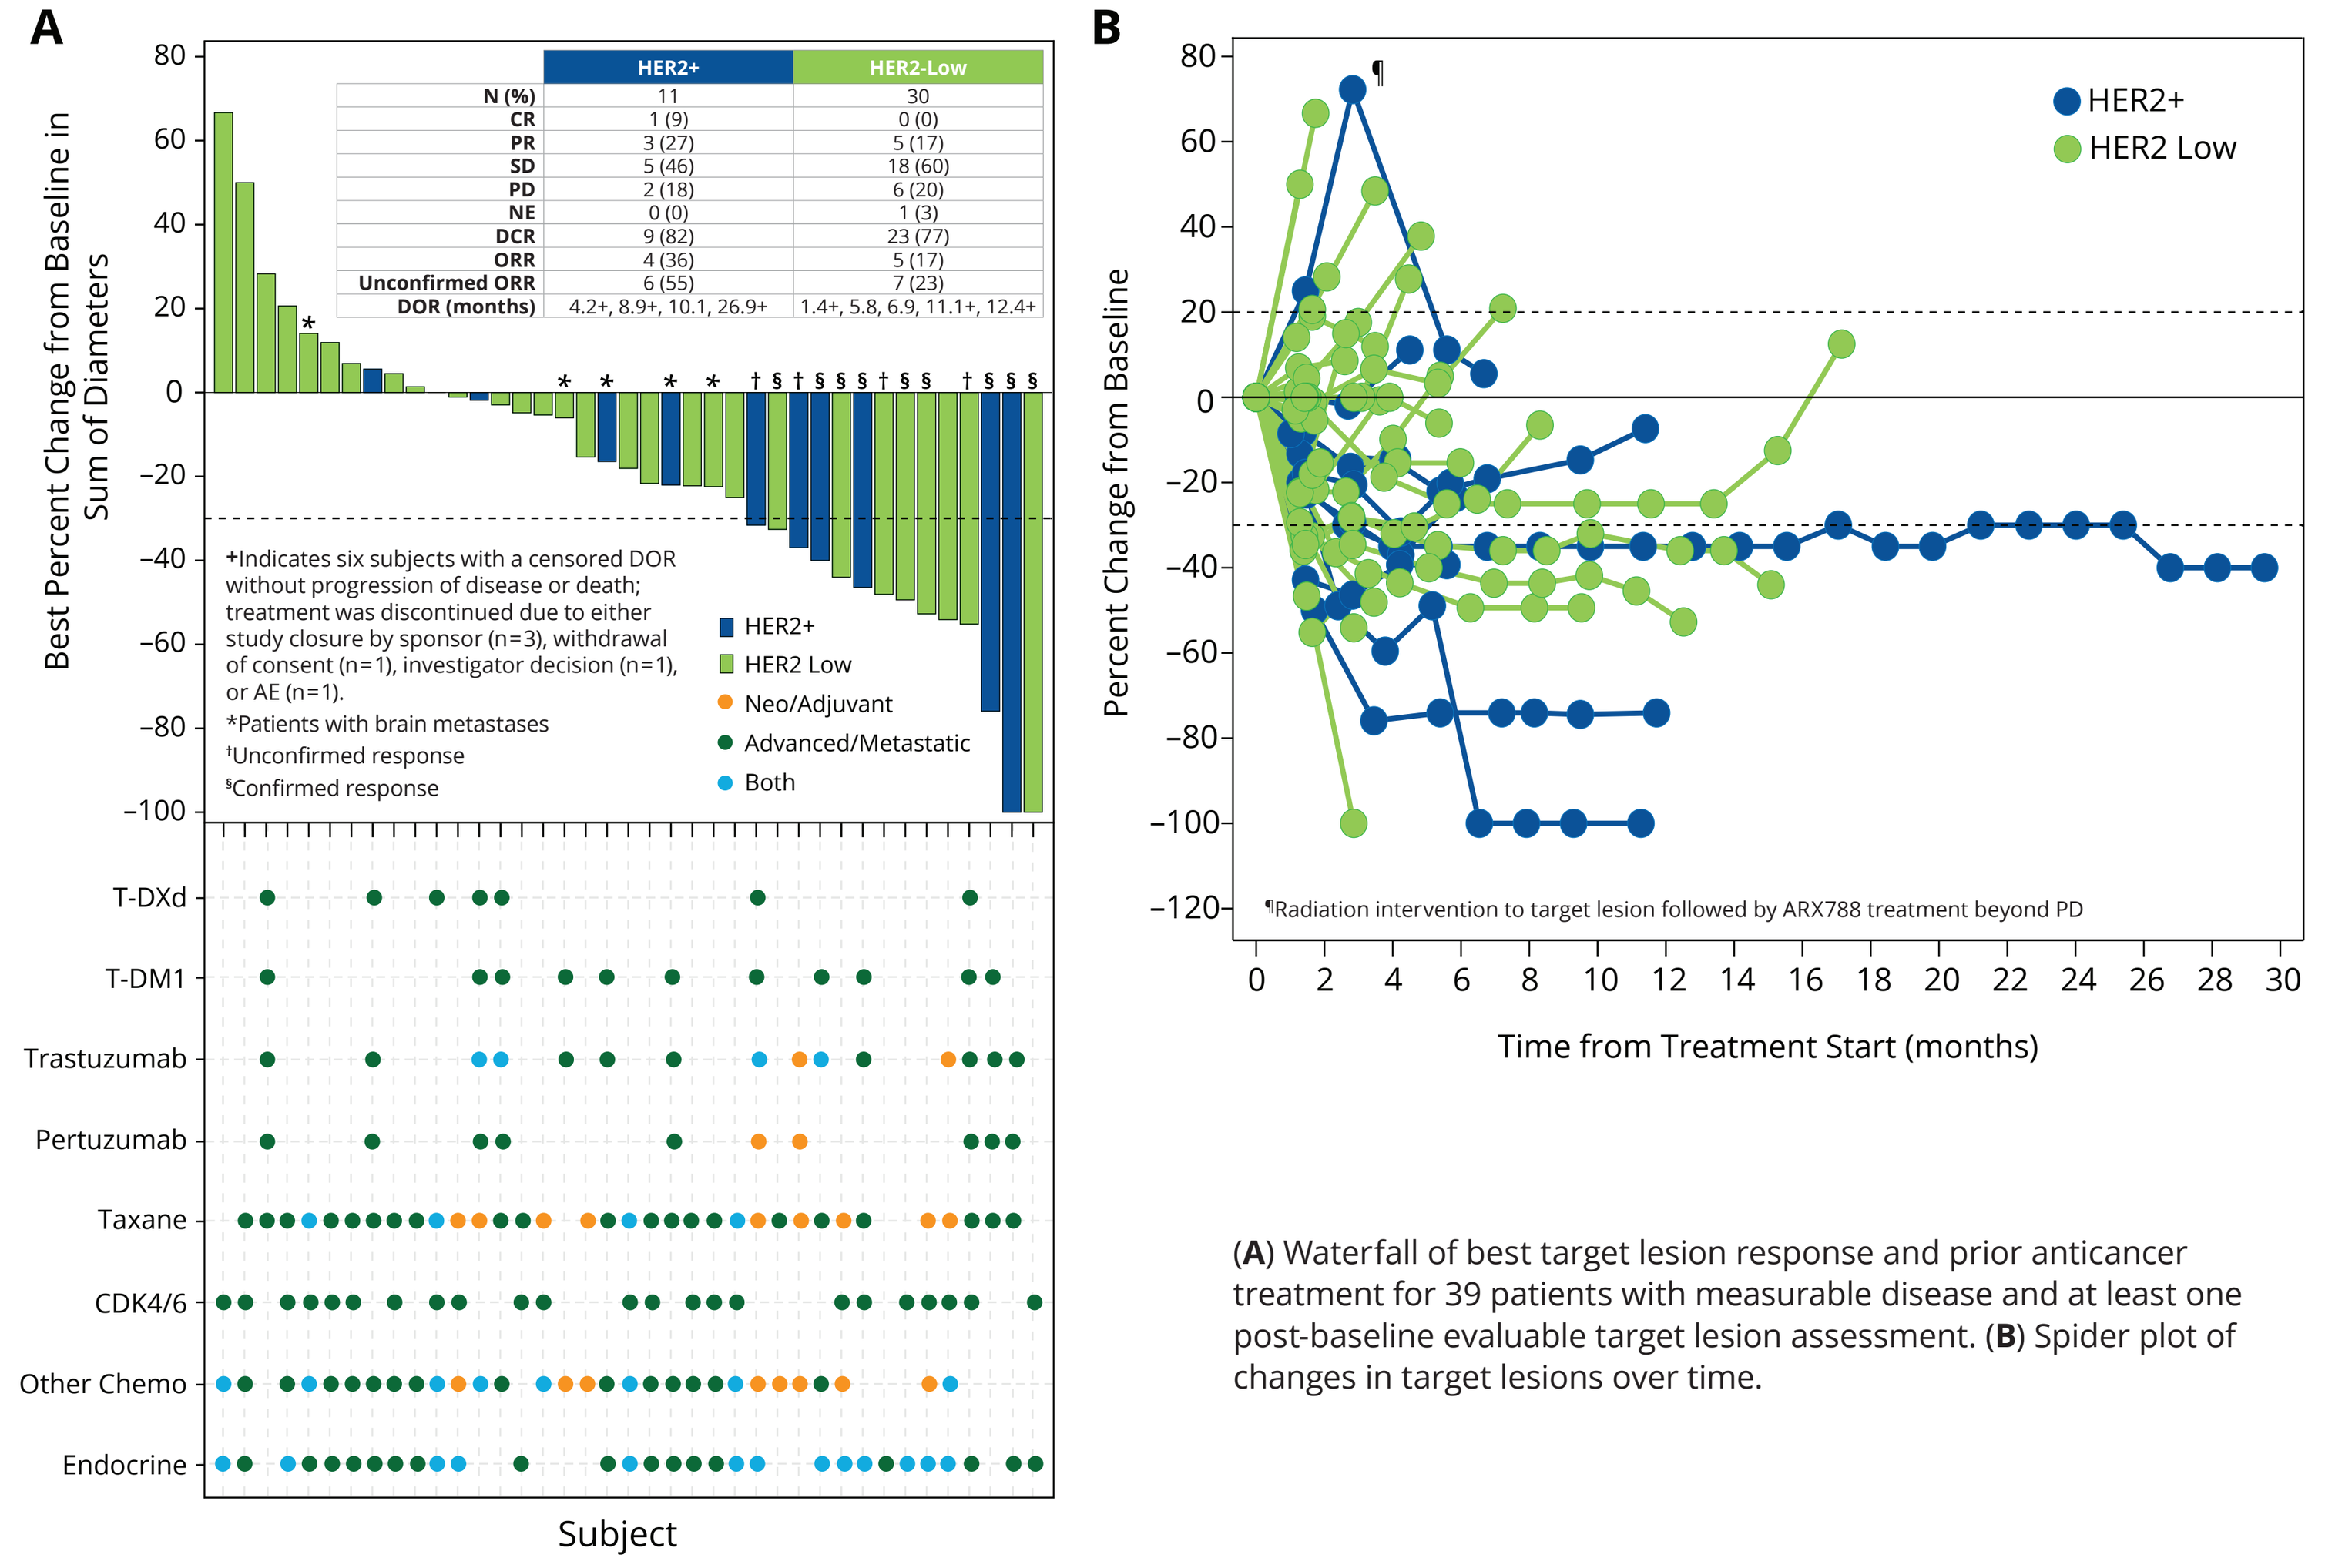
A
B
Best Percent Change from Baseline in Sum of Diameters
	HER2+	HER2-Low
N (%)	11	30
CR	1 (9)	0 (0)
PR	3 (27)	5 (17)
SD	5 (46)	18 (60)
PD	2 (18)	6 (20)
NE	0 (0)	1 (3)
DCR	9 (82)	23 (77)
ORR	4 (36)	5 (17)
Unconfirmed ORR	6 (55)	7 (23)
DOR (months)	4.2+, 8.9+, 10.1, 26.9+	1.4+, 5.8, 6.9, 11.1+, 12.4+
+Indicates six subjects with a censored DOR without progression of disease or death; treatment was discontinued due to either study closure by sponsor (n=3), withdrawal of consent (n=1), investigator decision (n=1), or AE (n=1).
*Patients with brain metastases
†Unconfirmed response
§Confirmed response
HER2+
HER2 Low
Neo/Adjuvant
Advanced/Metastatic
Both
T-DXd
T-DM1
Trastuzumab
Pertuzumab
Taxane
CDK4/6
Other Chemo
Endocrine
Subject
Percent Change from Baseline
HER2+
HER2 Low
¶Radiation intervention to target lesion followed by ARX788 treatment beyond PD
Time from Treatment Start (months)

(**A**) Waterfall of best target lesion response and prior anticancer treatment for 39 patients with measurable disease and at least one post-baseline evaluable target lesion assessment. (**B**) Spider plot of changes in target lesions over time.

PATIENT CASE STUDIES

Case 1: Durable response in heavily pretreated HER2+ breast cancer

67-year-old white female
HER2+ (IHC 2+/ISH+) / ER+PR- / Baseline disease: left lung and abdomen
Best RECIST v1.1 response: confirmed PR (35% target lesion reduction)

PR (Day 81) · PR (Day 123)

4 months | 60 months | 101 months | 30 months

Prior Neoadjuvant Rx	Prior Adjuvant Rx	Prior Metastatic Rx
5-FU, epirubicin, cyclophosphamide, docetaxel	trastuzumab, letrozole, anastrozole, tamoxifen	paclitaxel, trastuzumab, exemestane, *T-DM1*, fulvestrant, trastuzumab, paclitaxel, capecitabine, vinorelbine

→ ARX788 (1.5 mg/kg Q6W)

Case 2: Antitumor activity in breast cancer patient with prior TDXd

41-year-old white female
HER2+ (IHC 2+/ISH+) / ER+PR+ / Baseline disease: bilateral lungs, abdomen, peritoneum
Best RECIST v1.1 response: unconfirmed PR (32% target lesion reduction)

PR (Day 130)

3 months | 19 months | 28 months | 5 m

Prior Neoadjuvant Rx	Prior Adjuvant Rx	Prior Metastatic Rx
TCHP	anastrozole, letrozole	*T-DM1*, exemestane, trastuzumab, capecitabine, tucatinib, *T-DXd*

→ ARX788 (1.6 mg/kg Q3W)

CONCLUSIONS

- **ARX788 demonstrated a favorable and differentiated safety profile compared with other HER2-targeted ADCs.**
 - **TRAEs were mostly low Grade; Grade 3 events occurred at a low frequency (<25%), and there were no Grade 4 or 5 TRAEs. This compares favorably to T-DXd and T-DM1, which have reported ≥40% incidence of Grade 3 or 4 TRAEs.[3]**
 - **Cytopenias were infrequent and no ≥Grade 3 events were observed. Such AEs can be dose limiting with other HER2-targeted ADCs.[3,4,6-8]**
 - **Ocular toxicity was predominantly low-grade and reversible.**
- **Antitumor activity was seen with ARX788 in heavily pretreated HER2+ and HER2-low breast cancer, including tumor reductions in patients with prior T-DXd exposure.**
- **The ACE-Breast-03 study (NCT04829604) is currently evaluating ARX788 in patients with HER2+ advanced breast cancer previously treated with T-DXd. Enrollment is now open [see poster PO1-04-02].**

Appreciation

We thank the patients who have enrolled in the ACE-PanTumor-01 trial. We appreciate the hard work of our investigators andsite staff who have contributed their knowledge and expertise in the design and conduct of this trial.

References

1. Zhang J et al. Phase I Trial of a Novel Anti-HER2 Antibody-Drug Conjugate, ARX788, for the Treatment of HER2-Positive Metastatic Breast Cancer. *Clin Cancer Res.* 2022;28:4212-21.; **2.** Hurvitz S et al. Safety and unique pharmacokinetic profile ofARX788, a site-specific ADC, in heavily pretreated patients with HER2-overexpresing solid tumors: Results from two phase 1 clinical trials. *JCO* 2021; suppl.1038.; **3.** Cortes J et al. Trastuzumab Deruxtecan versus Trastuzumab Emtansine for Breast Cancer. *NEJM* 2022;386:1143-54.; **4.** Modi S et al. Trastuzumab Deruxtecan in Previously Treated HER2-Low Advanced Breast Cancer. *NEJM* 2022;387:9-20.; **5.** Rosa K. ARX788 Meets PFS End Point in HER2+ Metastatic Breast Cancer. OncLive.com 2023.; **6.** Verma S et al. Trastuzumab Emtansine for HER2-Positive Advanced Breast Cancer. *NEJM* 2012;367(19):1783-91.; **7.** Modi S et al. Trastuzumab Deruxtecan in Previously Treated HER2-Positive Breast Cancer. *NEJM* 2020;382(7):610-21.; **8.** Krop IE et al. Trastuzumab emtansine versus treatment of physician's choice for pretreated HER2-positive advanced breast cancer (TH3RESA): A randomised, open-label, phase 3 trial. *Lancet Oncol.* 2014;15:689–699.

Poster contact
breast03trialinquiry@ambrx.com

Copies of this poster obtained through Quick Response (QR) Code are for personal use only and may not be reproduced without permission from SABCS® and the author of this poster.

PRESENTED AT THE 46TH ANNUAL SAN ANTONIO BREAST CANCER SYMPOSIUM (SABCS); DECEMBER 5–9, 2023; SAN ANTONIO, TX.